Combination of the aptamer AS1411 with paclitaxel or Ara-C produces synergistic inhibition of cancer cell growth

Abstract No.: 4818

Christy Ritchie, Benjamin Doran, Kirti Shah, Gail Rowlinson-Busza, Nigel Courtenay-Luck, David Jones • Antisoma Research Ltd., St.George's Hospital Medical School, London, UK

Introduction

- Aptamers are oligonucleotides with a defined tertiary structure, which enables them to bind to specific cellular proteins.[1] AS1411 is a 26-base G-rich oligonucleotide which forms a dimeric G-quadruplex structure that is resistant to degradation by nucleases[2,3]
- AS1411 binds to the protein nucleolin, which is expressed at elevated levels in transformed cells. Tumor cells have been shown to present nucleolin on the cell surface as well as expressing it in the cytoplasm and nucleus. Nucleolin plays multiple roles in the cell and is involved in ribosome biogenesis, cell growth and DNA replication
- A phase I trial of AS1411 in 30 advanced cancer patients showed promising signs of activity, including two objective responses among 12 of these patients with metastatic renal cancer. No serious toxicity of any type related to drug administration was observed[4]
- The studies described here investigate the mechanism of cell killing induced by AS1411 and its effect when used in combination with standard cytotoxic drugs such as paclitaxel and Ara-C

Methods

- **Sulphorhodamine B assay.** Cells were seeded in wells of a 96-well plate at a number optimized for each cell line. AS1411 was added and cells were incubated for 6 days (or as described in the experiment). Cells were then washed, fixed to the 96-well plate and exposed to the dye SRB. Remaining cell mass was measured with a spectrophotometer and IC_{50} determined
- **Apoptosis assays.** DNA fragmentation was measured using the Cell Death Detection ELISA Plus (Roche Applied Science). Caspase activation was measured using APO-ONE Homogeneous Caspase 3/7 Assay (Promega). Alternatively, caspase activation was measured on a Guava cell analyzer using Guava multi-caspase kit (Guava Technologies). Phosphatidylserine (PS) externalization was measured using Guava Nexin kit (Guava Technologies)
- **Cell cycle analysis.** Cells were analyzed on a Guava cell analyzer using Guava cell cycle reagent (Guava Technologies)

Results

Breadth of tumor cell killing

Table 1: IC_{50} values for various cell lines treated with AS1411 as measured by Sulphorhodamine B assay

Cell line	Tumor type	Average IC_{50} µM
DU-145	Prostate	4.9
PC3	Prostate	5.2
A549	Lung	5.9
CALU-6	Lung	10.6
LS174T	Colon	5.1
HT-29	Colon	8.8
MCF-7	Breast	4.2
ZR75-1	Breast	7.9
HGC.27	Gastric	11.2
KATO III	Gastric	26.5
A498	Renal	3.1
PANC-1	Pancreatic	4.6
SKOV-3	Ovarian	7.9
U87-MG	Glioblastoma	1.9
SK-MEL-28	Melanoma	5.4
MV4-11	AML	1.9
HS 27	Fibroblast	No kill

Figure 1: Cell kill as measured by Sulphorhodamine B assay

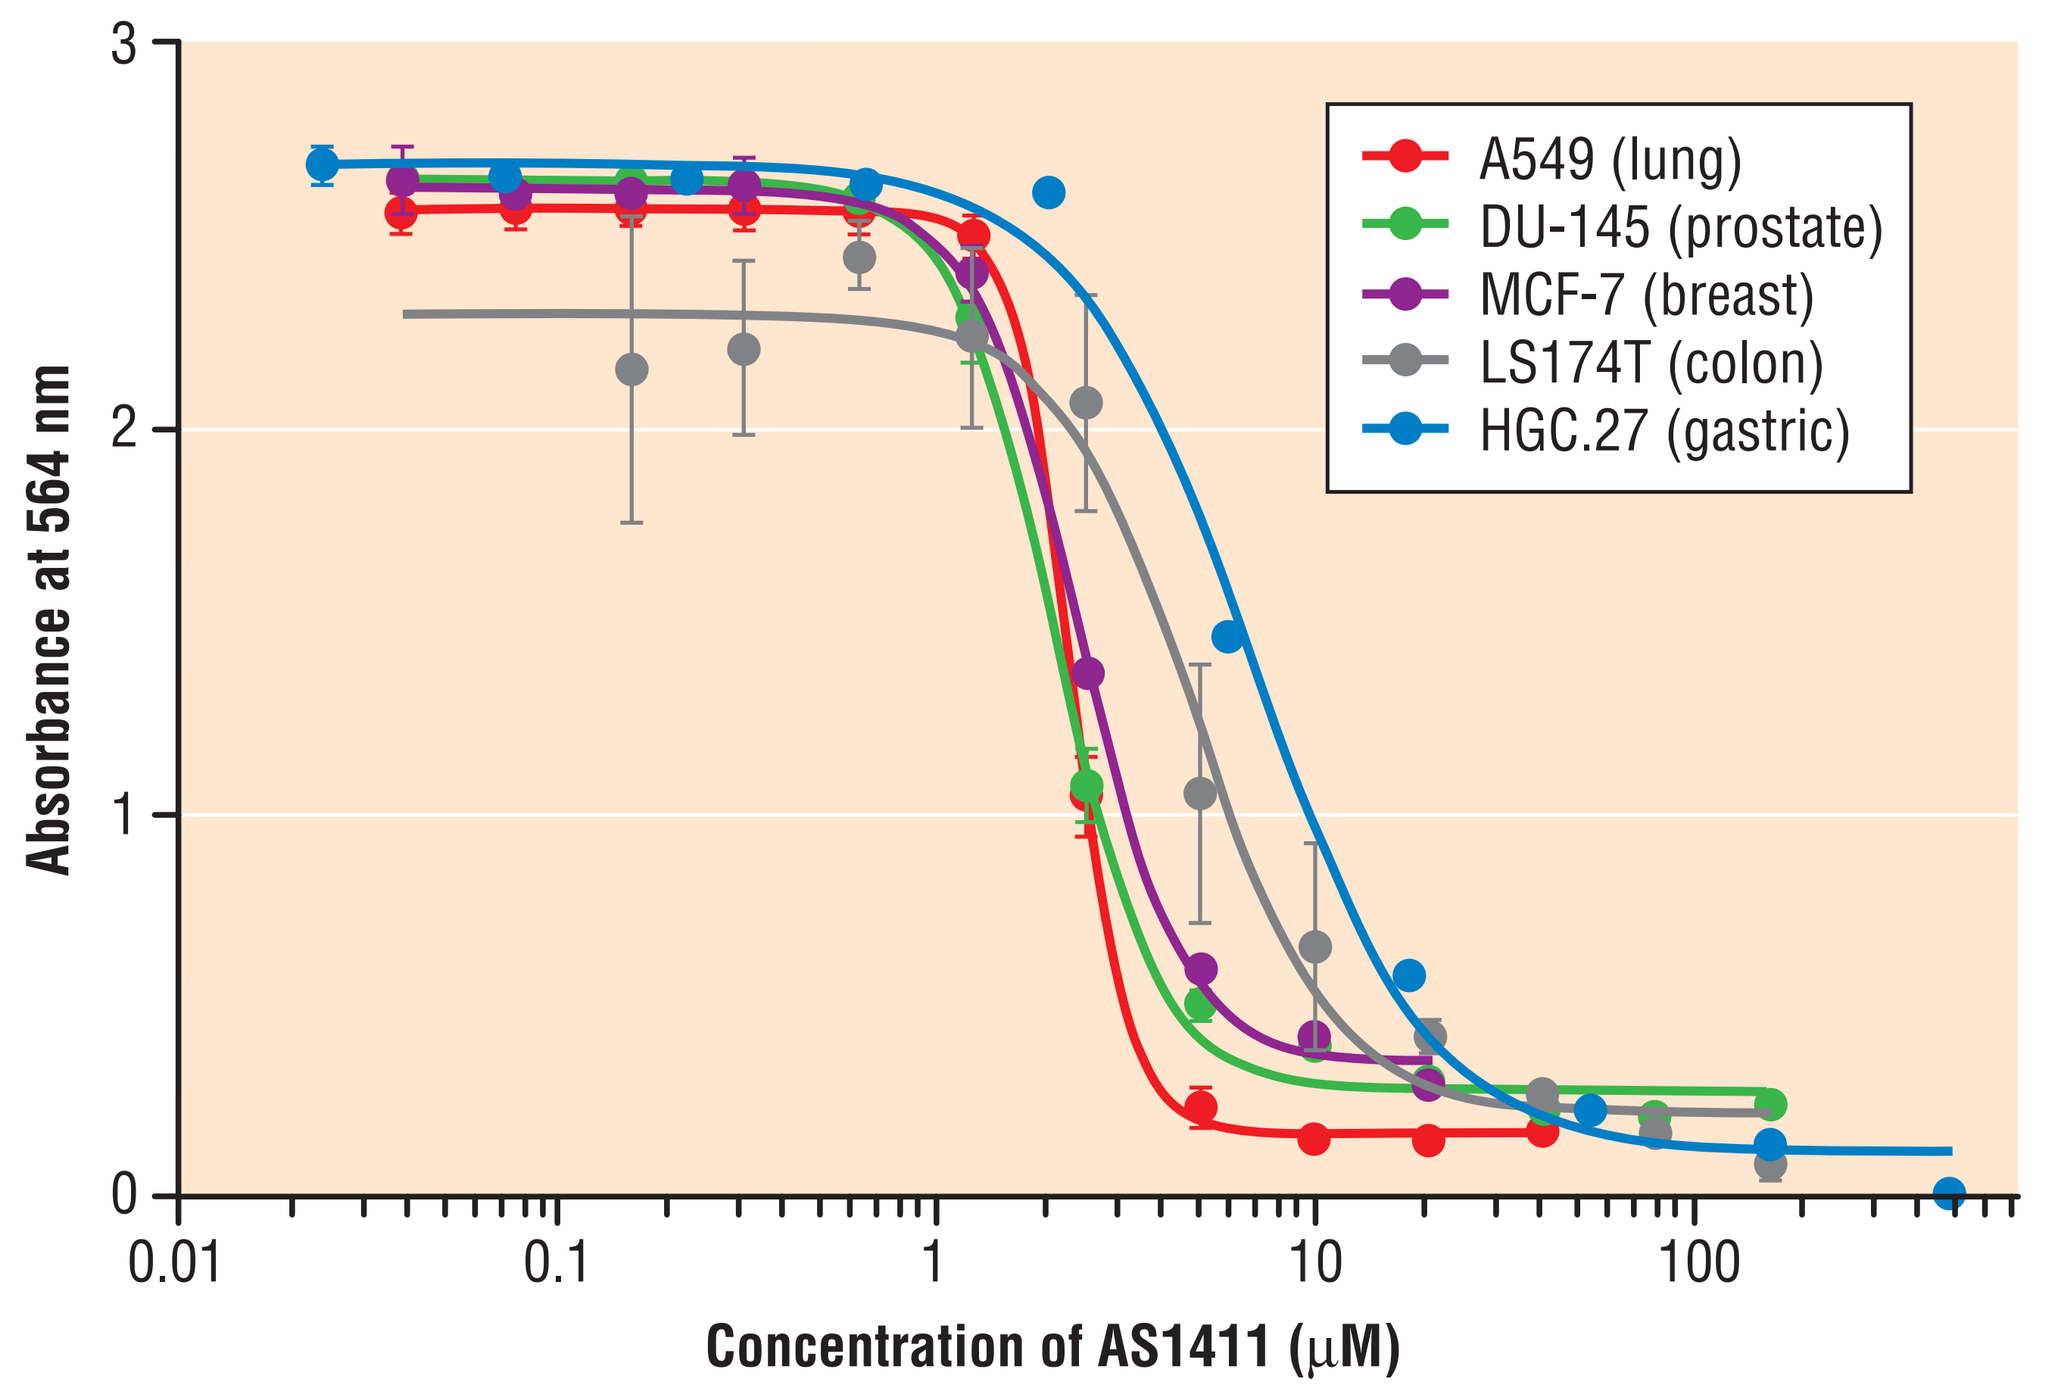
A549 (lung)
DU-145 (prostate)
MCF-7 (breast)
LS174T (colon)
HGC.27 (gastric)
Absorbance at 564 nm
Concentration of AS1411 (µM)

Tumor cells were grown in the presence of AS1411 in a 6-day assay. The table shows that all tumor cell lines tested to date are sensitive to AS1411, with IC_{50} values typically in the range 2–10 µM. Normal fibroblast cells were unaffected by similar concentrations of AS1411.

The graph shows data from several selected cell lines.

Mechanism of tumor cell killing

Figure 2a: Evidence of apoptotic cell death: caspase activation

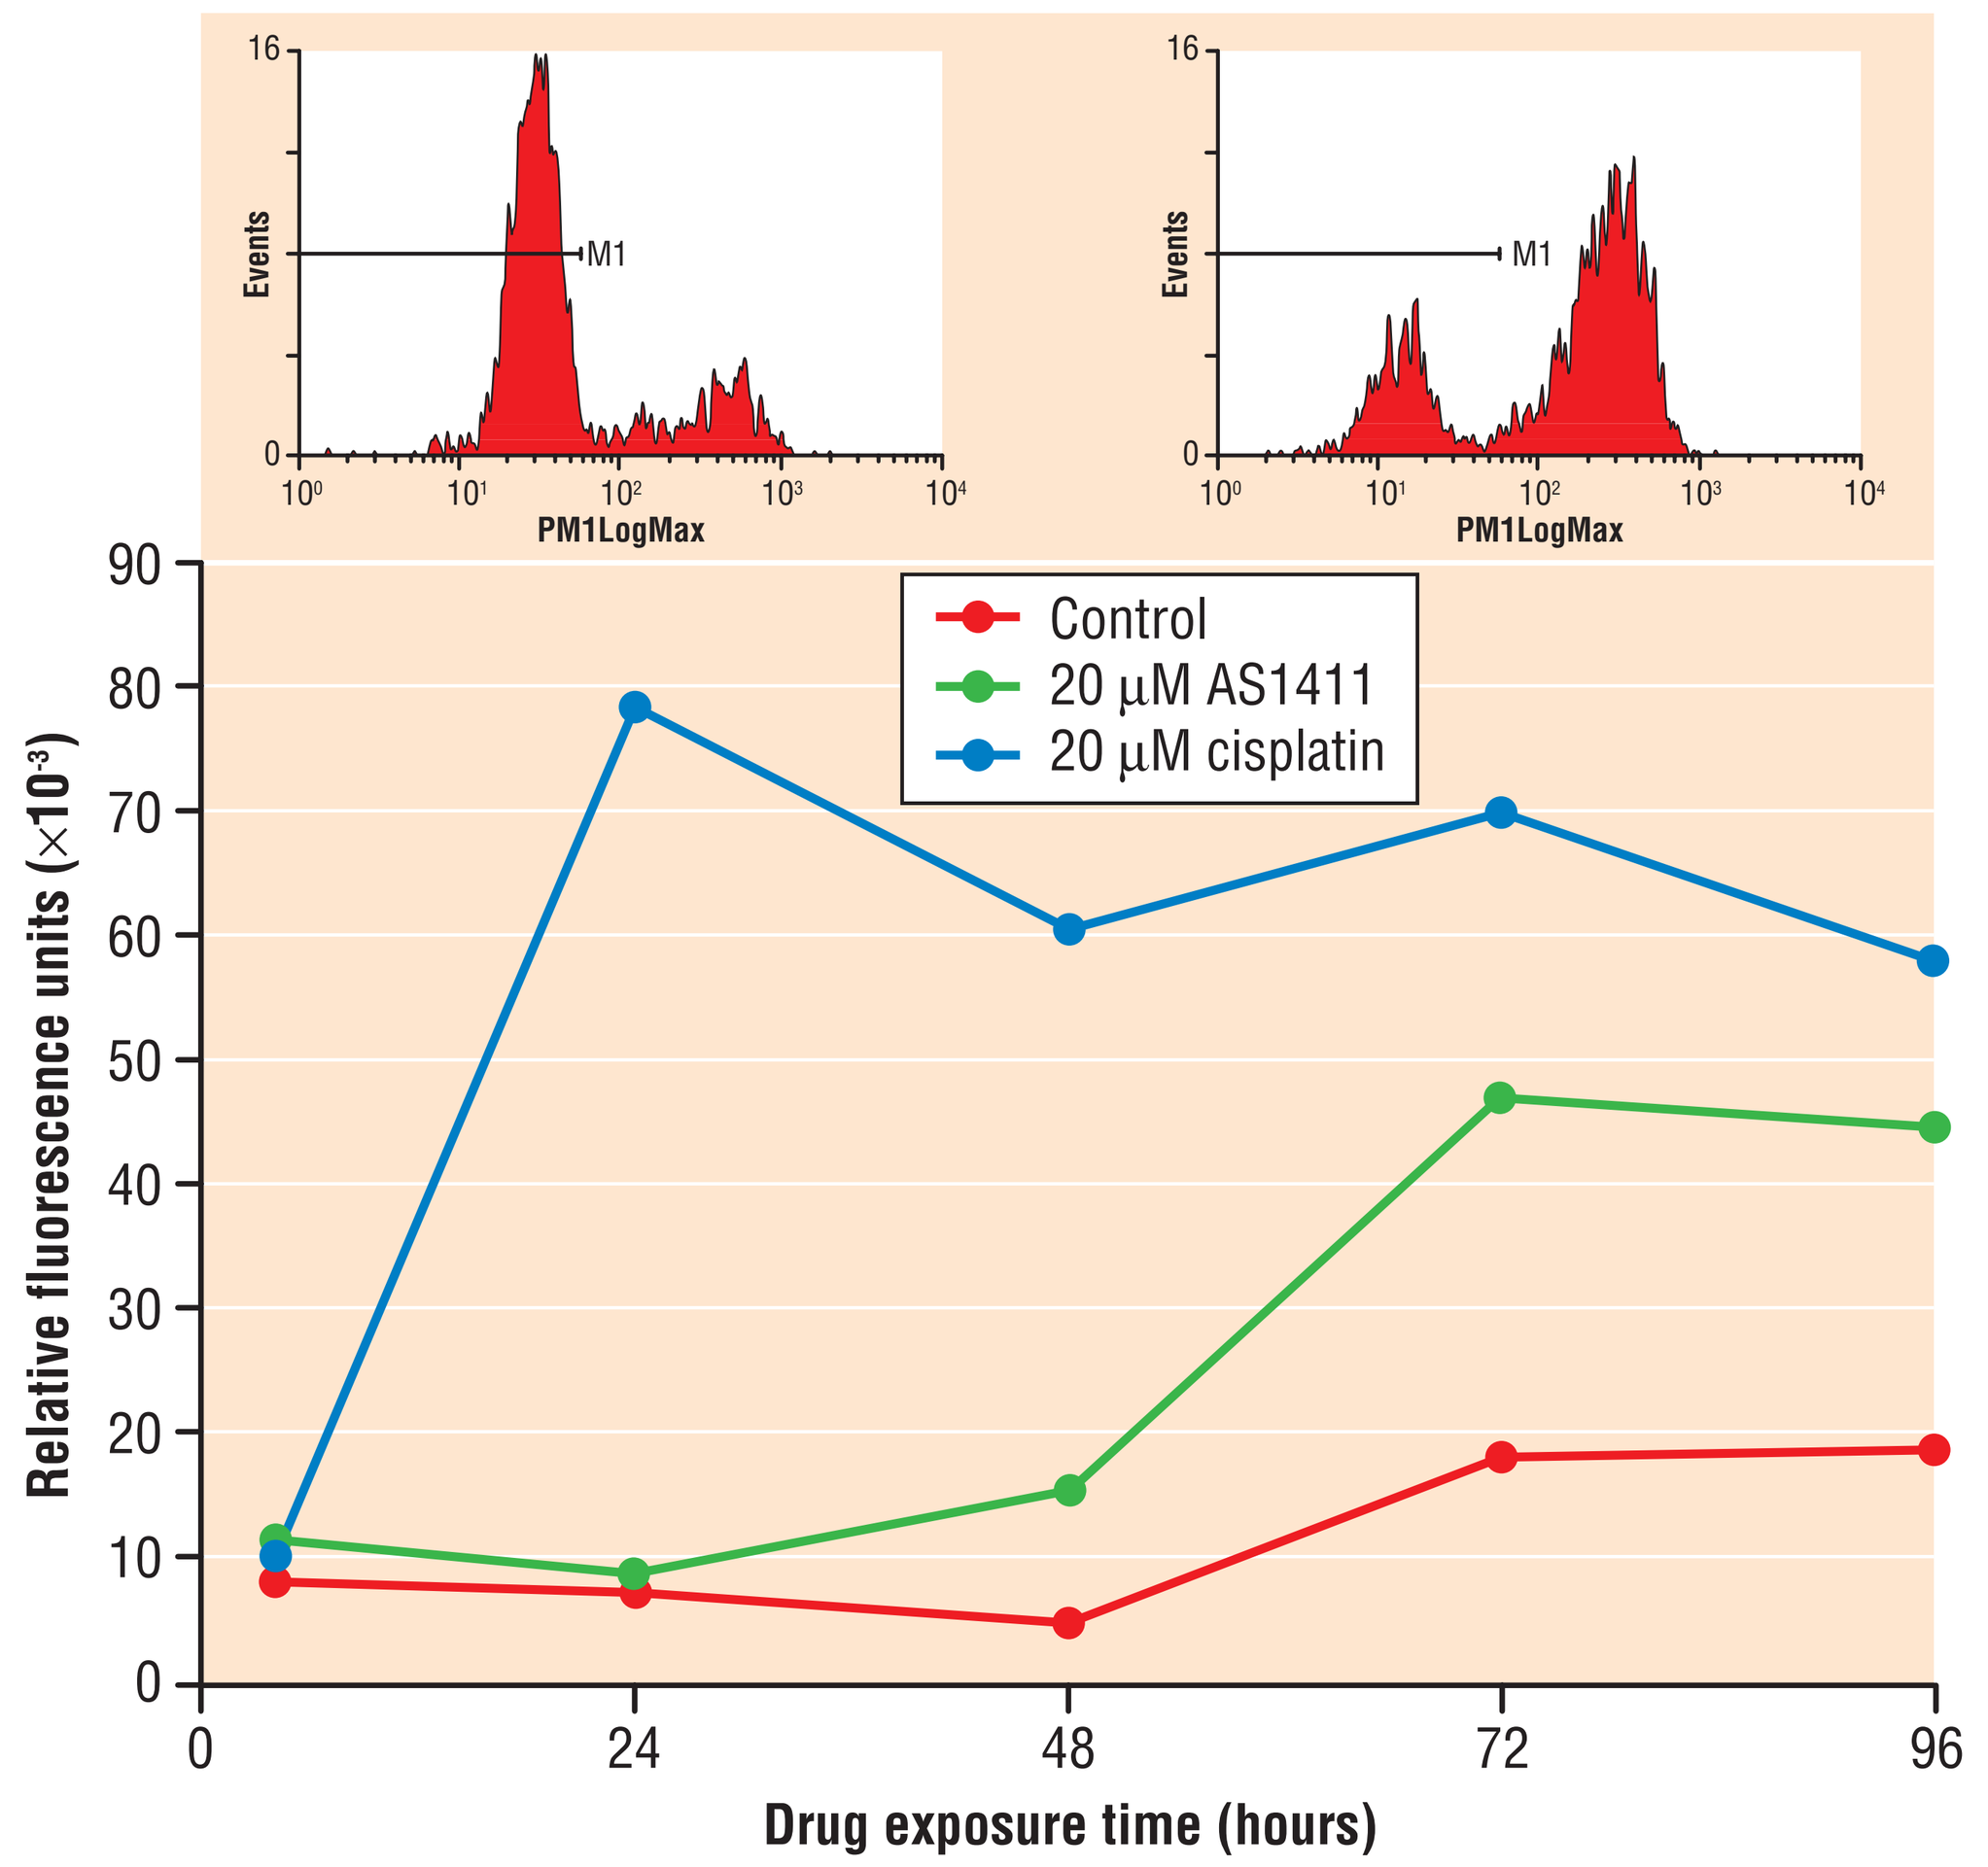
Events
M1
PM1LogMax
Control
20 µM AS1411
20 µM cisplatin
Relative fluorescence units (×10⁻³)
Drug exposure time (hours)

The graph shows MV4-11 (AML) cells treated with 20 µM AS1411 or cisplatin. Caspase 3/7 activity increased rapidly upon treatment with cisplatin, but only began to increase after 48 to 72 hours when cells were incubated with AS1411.

Cell cytometric analysis (inset); left plot: control cells; right plot: AS1411 treated cells. At 72 hours, around 72% of AS1411 treated cells had elevated levels of activated caspases (as shown by right-hand peak).

Figure 2b: Evidence of apoptotic cell death: PS externalization

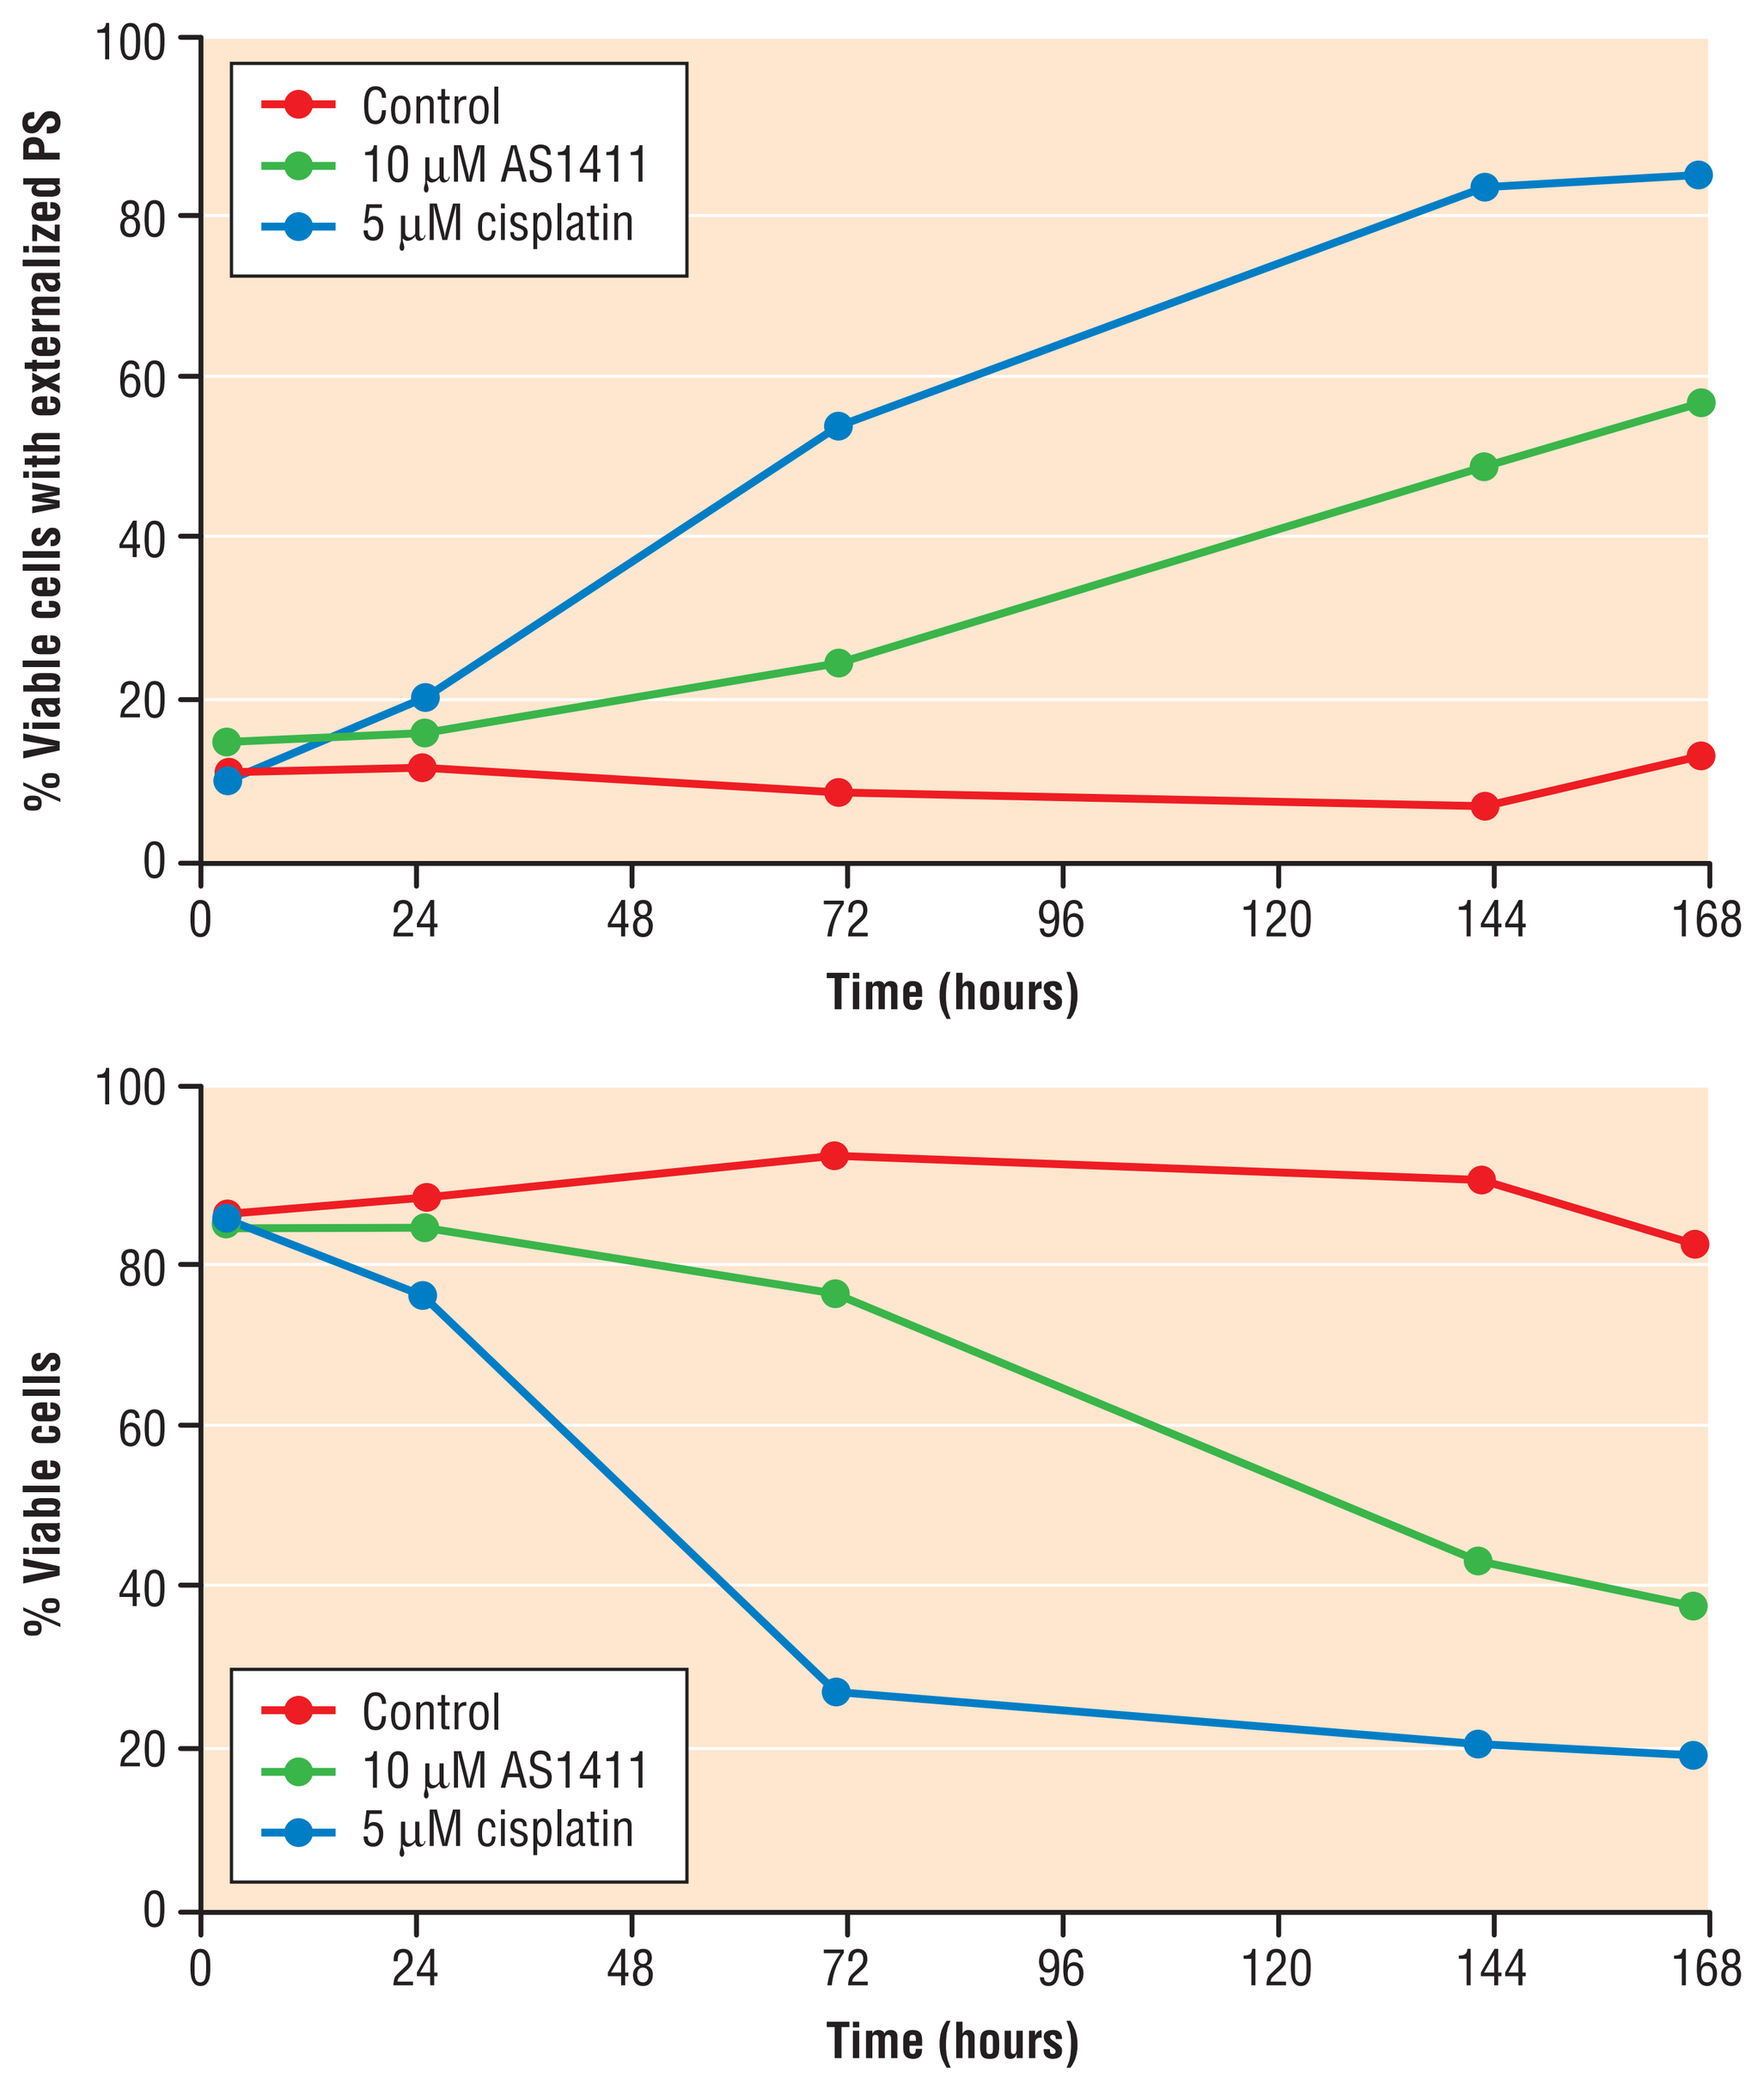
Control
10 µM AS1411
5 µM cisplatin
% Viable cells with externalized PS
% Viable cells
Time (hours)

Phosphatidylserine (PS) externalization was measured by cell cytometric analysis in MV4-11 cells treated with 10 µM AS1411 or 5 µM cisplatin. PS appears in the outer leaflet of the membrane 4 to 5 days after AS1411 treatment (upper graph), and this correlates with decreased cell viability measured in the same experiment by cell cytometric analysis (lower graph). Onset of PS externalization is slower with AS1411 than with cisplatin.

Figure 2c: Evidence of apoptotic cell death: DNA fragmentation

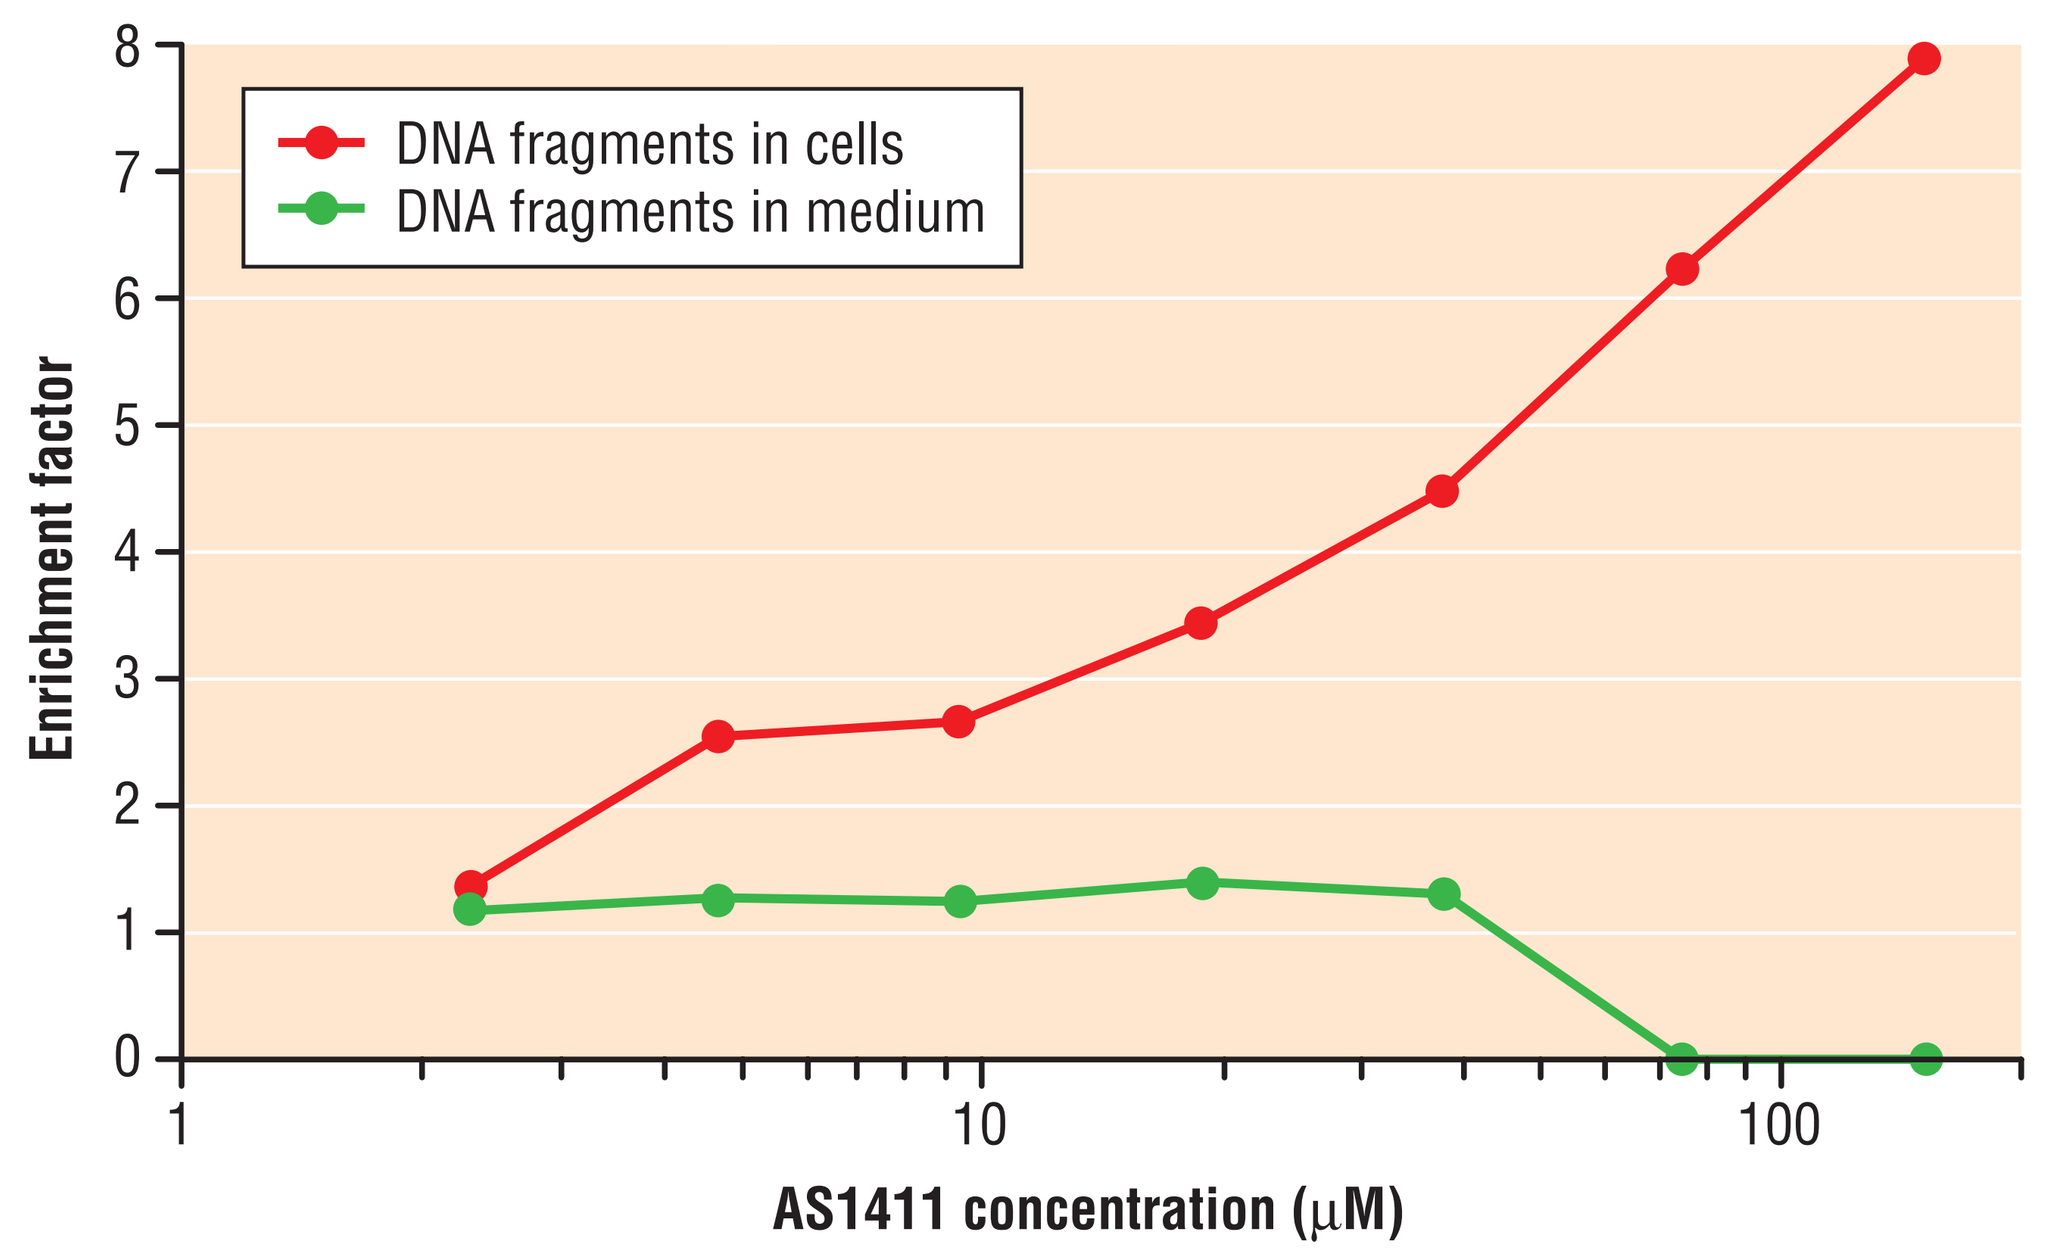
DNA fragments in cells
DNA fragments in medium
Enrichment factor
AS1411 concentration (µM)

DNA fragmentation was measured in DU-145 cells treated with increasing concentrations of AS1411 for a 4-day incubation. Fragmentation increased in a dose-dependent manner.

Dynamics of tumor cell killing

Figure 3: Cell growth and viability with/without AS1411

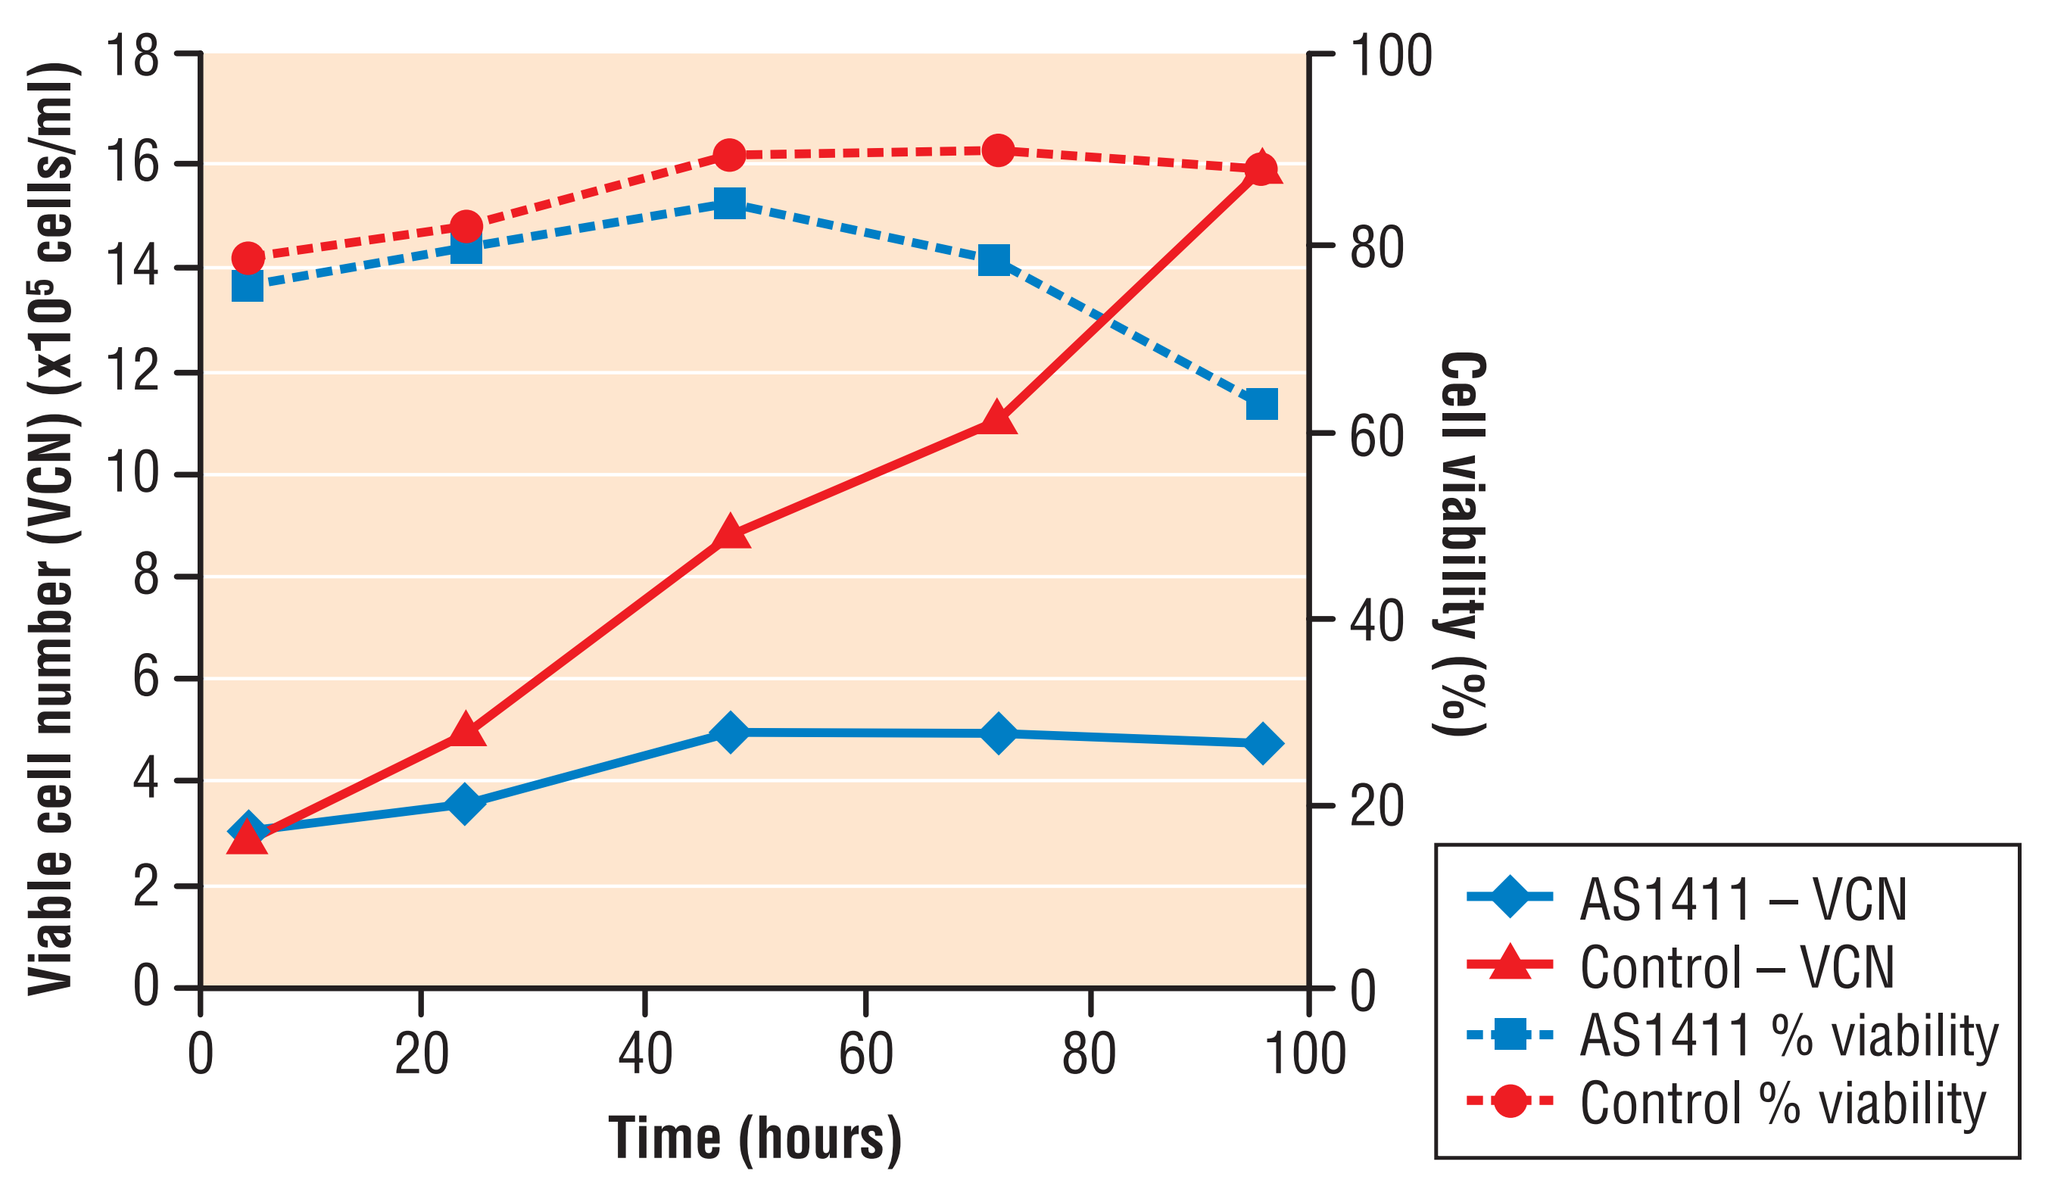
AS1411 – VCN
Control – VCN
AS1411 % viability
Control % viability
Viable cell number (VCN) (x10⁵ cells/ml)
Cell viability (%)
Time (hours)

Growth of DU-145 (prostate) cells is slowed in the presence of 20 µM AS1411. Viability remains high until 72 hours, after which cell viability drops.

Figure 4: Inhibition of cell growth and S phase arrest

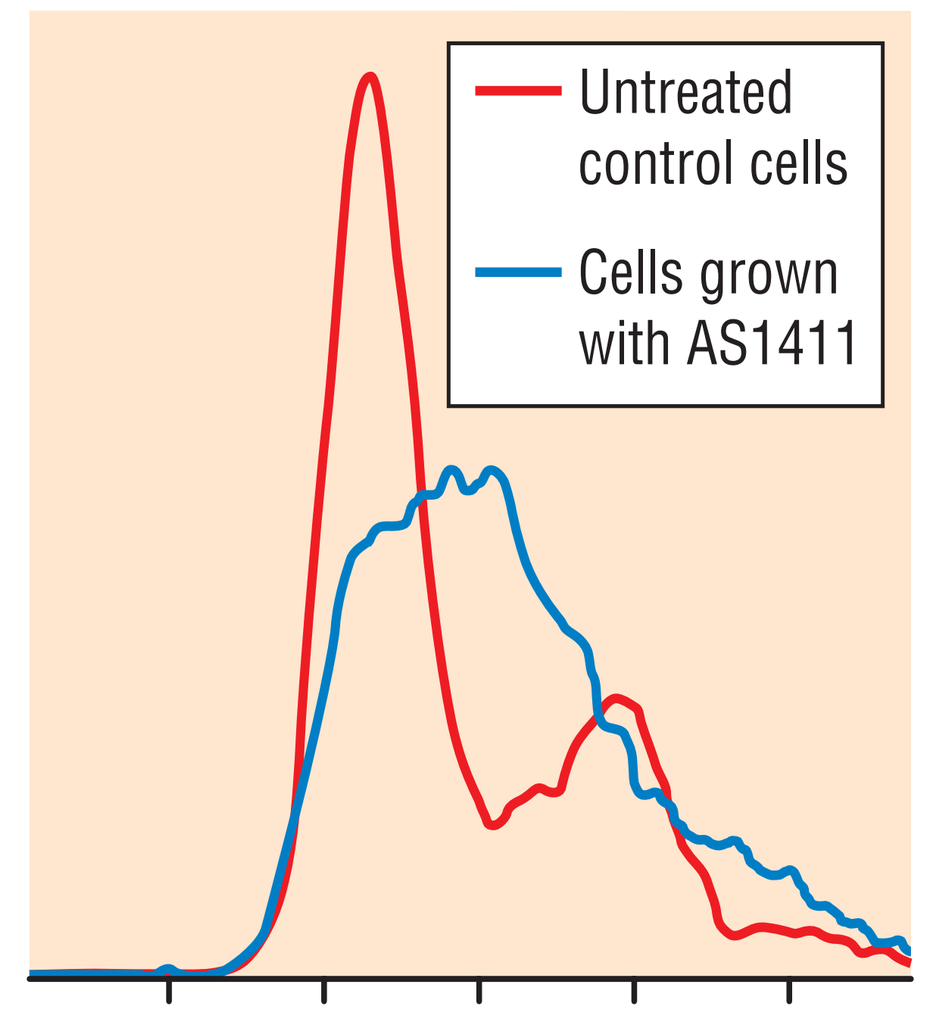
Untreated control cells
Cells grown with AS1411

Cell cytometric analysis of DNA content shows that DU-145 cells accumulate in S-phase when treated with 20 µM AS1411 for 48 hours.

For control cells the first (major) peak represents cells in G1, the second (minor) peak represents cells in G2; the region between represents cells in S phase.

Figure 5: Time course of cell kill

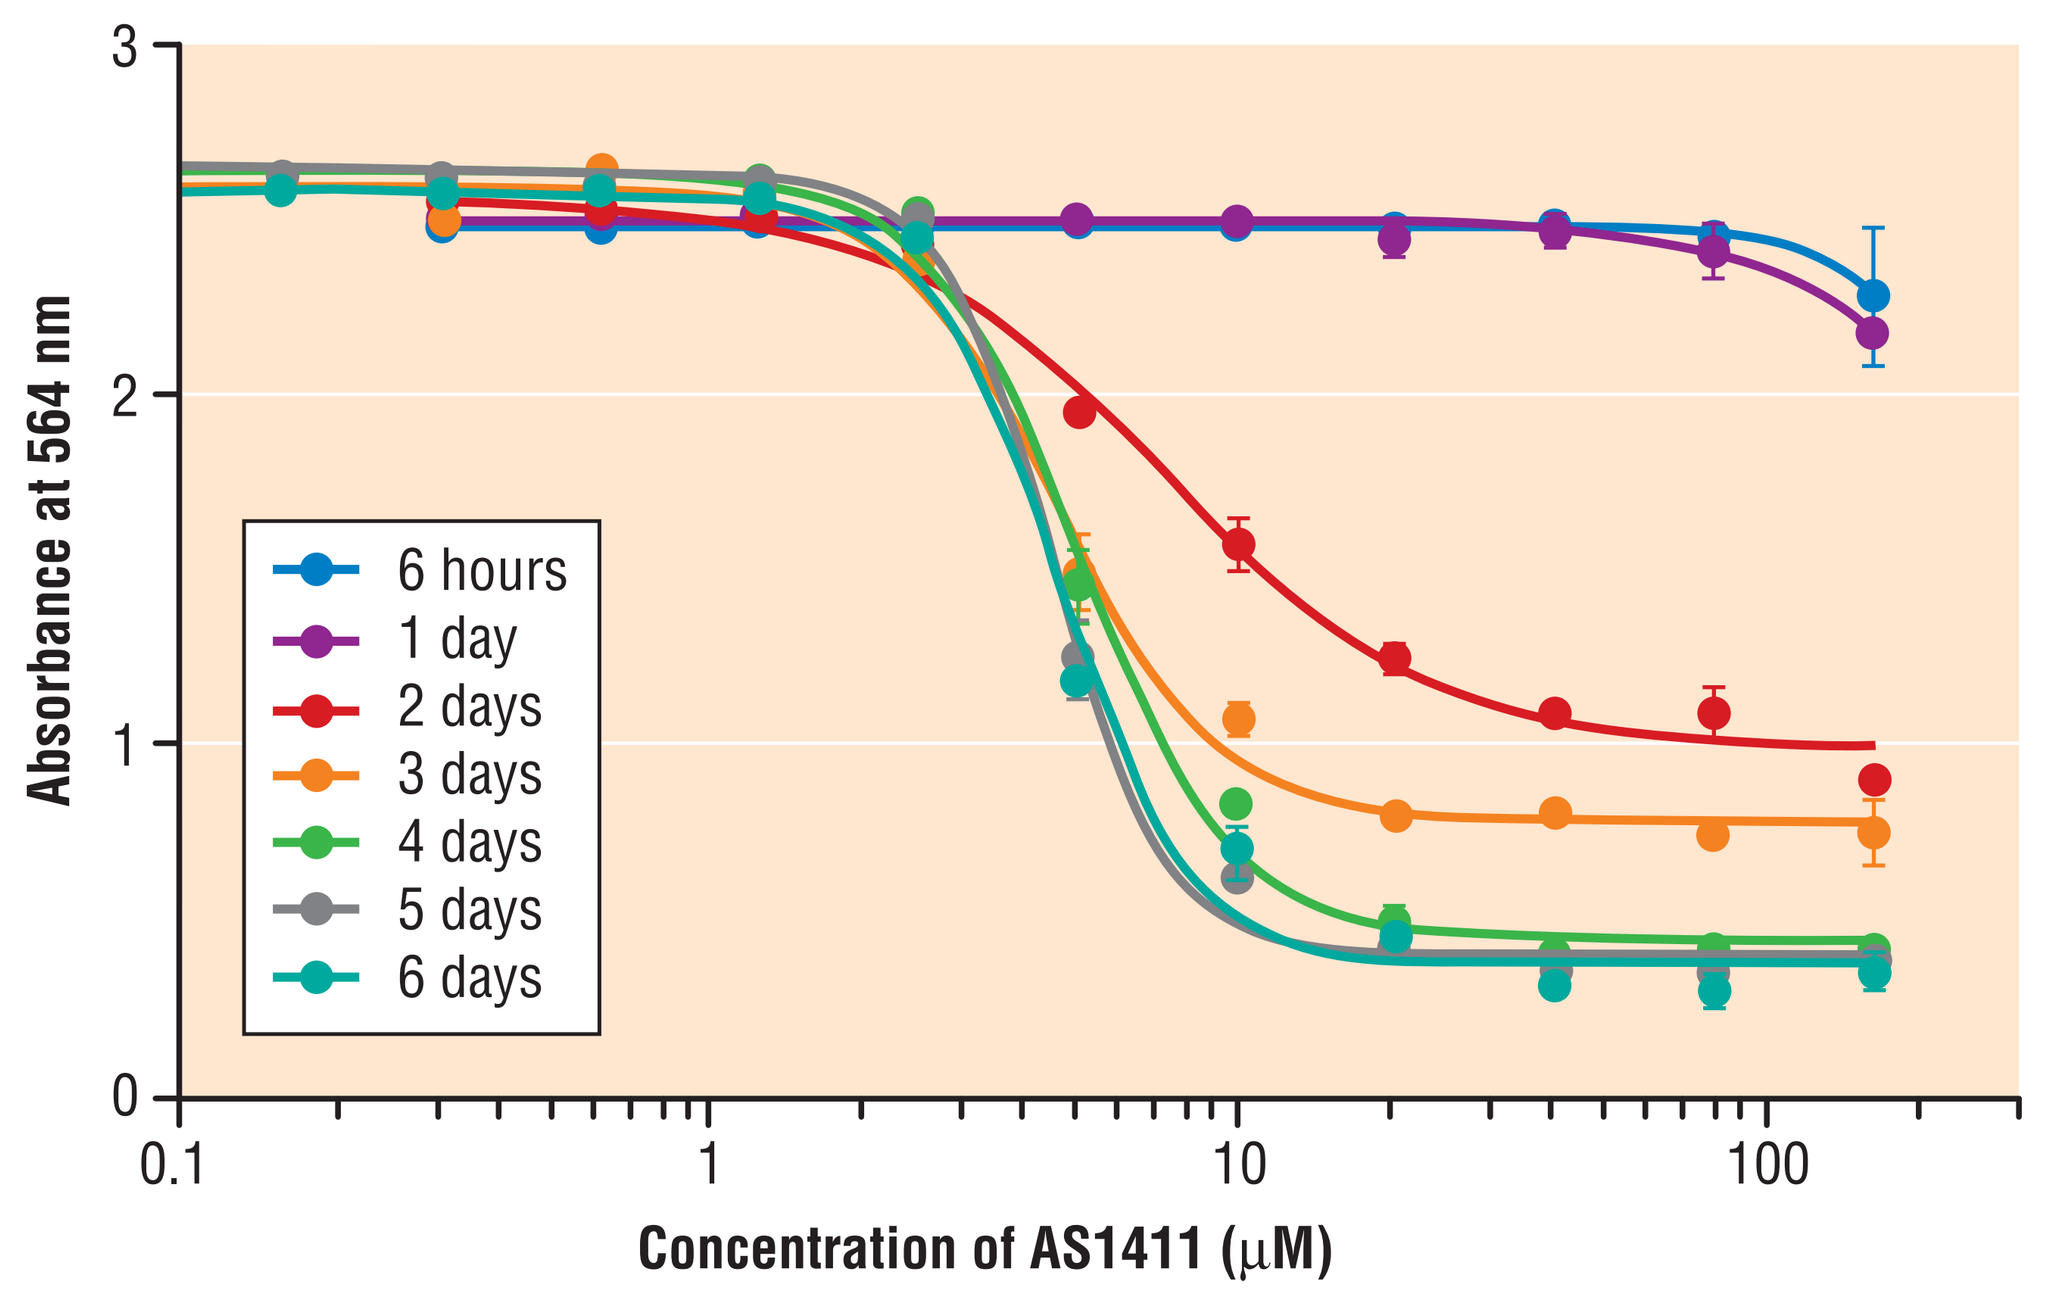
6 hours
1 day
2 days
3 days
4 days
5 days
6 days
Absorbance at 564 nm
Concentration of AS1411 (µM)

DU-145 cells were incubated with AS1411 for between 6 hours and 6 days, then drug was washed off the cells, fresh medium applied, and the cells left to grow for the full 6-day assay. For efficient cell kill, cells required exposure for 4 days or longer.

Synergistic effects with paclitaxel and Ara-C

Figure 6a: Combination of AS1411 with paclitaxel in the SRB cell kill assay with A549 cells

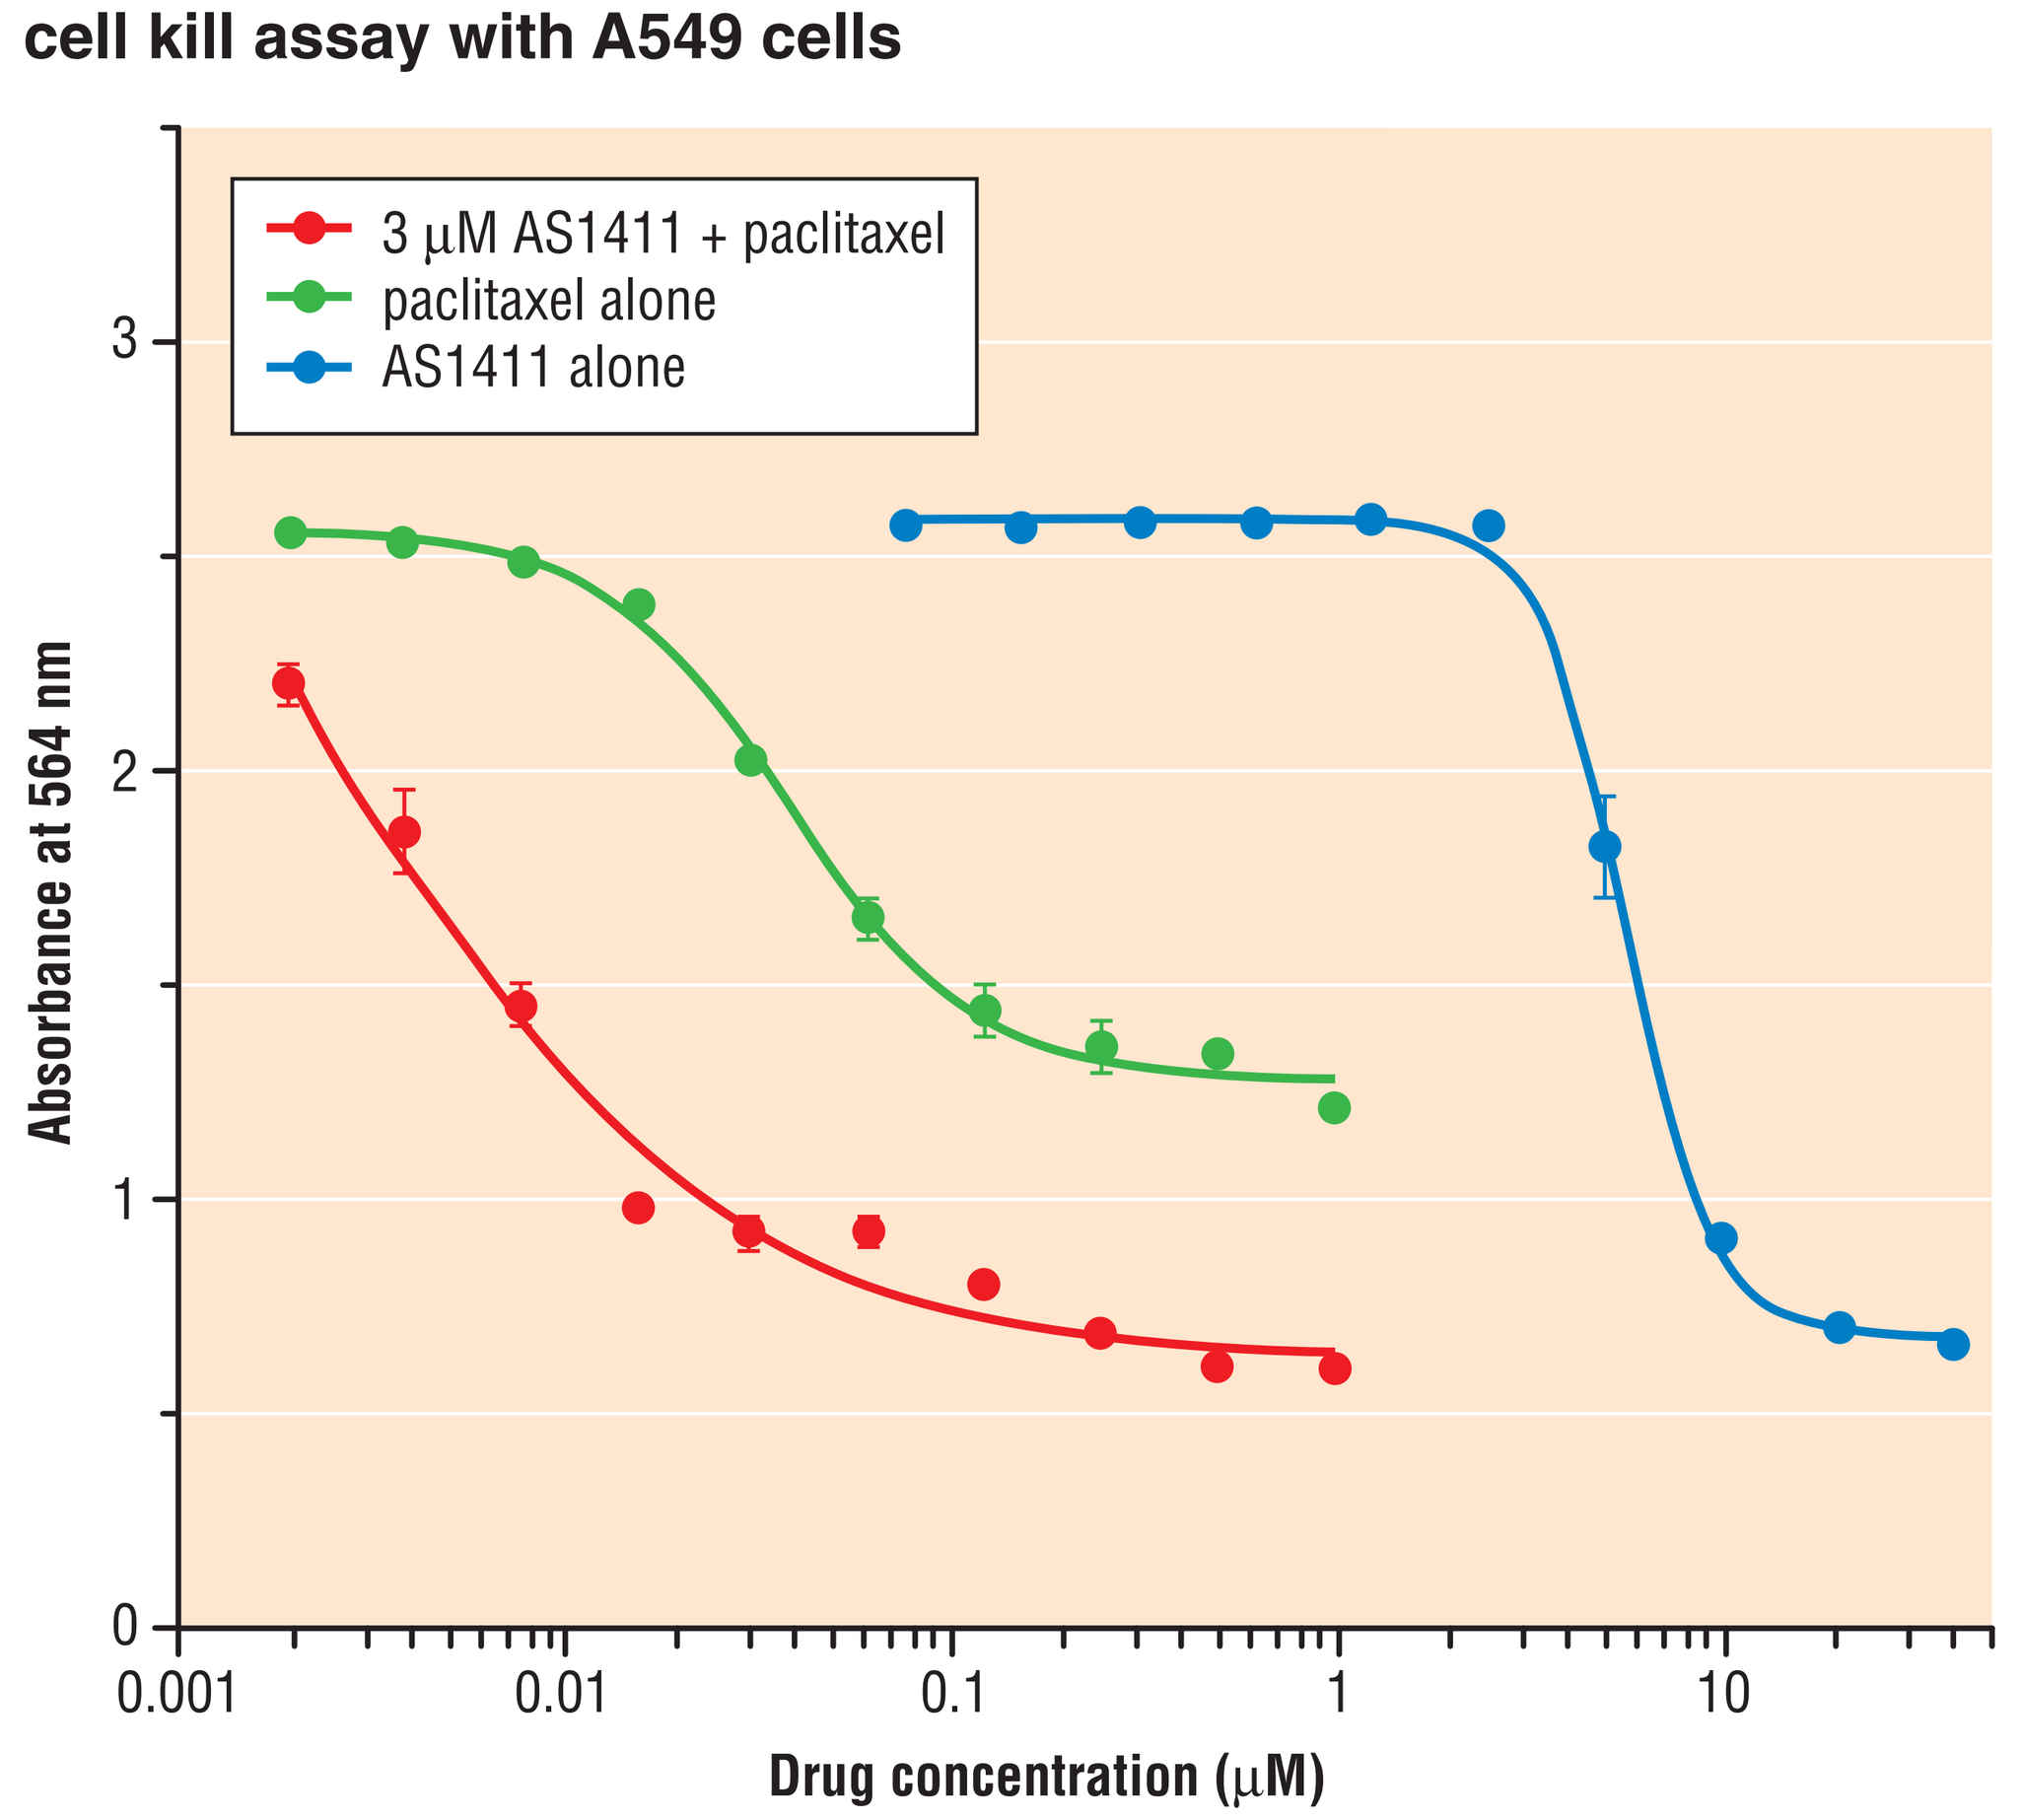
3 µM AS1411 + paclitaxel
paclitaxel alone
AS1411 alone
Absorbance at 564 nm
Drug concentration (µM)

Cells were incubated with a non-toxic (3 µM) concentration of AS1411 for the full 6-day assay. (Dose-related cell kill with AS1411 shown in blue). At day 3, paclitaxel was added at the concentrations shown. Synergistic cell kill activity is observed in both A549 (lung) and MCF-7 (breast; data not shown) cell lines as determined by combination index analysis. Synergy was not observed in this assay with cell lines HT-29, A498, PANC-1 or PC3.

Fig 6b: Combination of AS1411 with Ara-C with MV4-11 cells

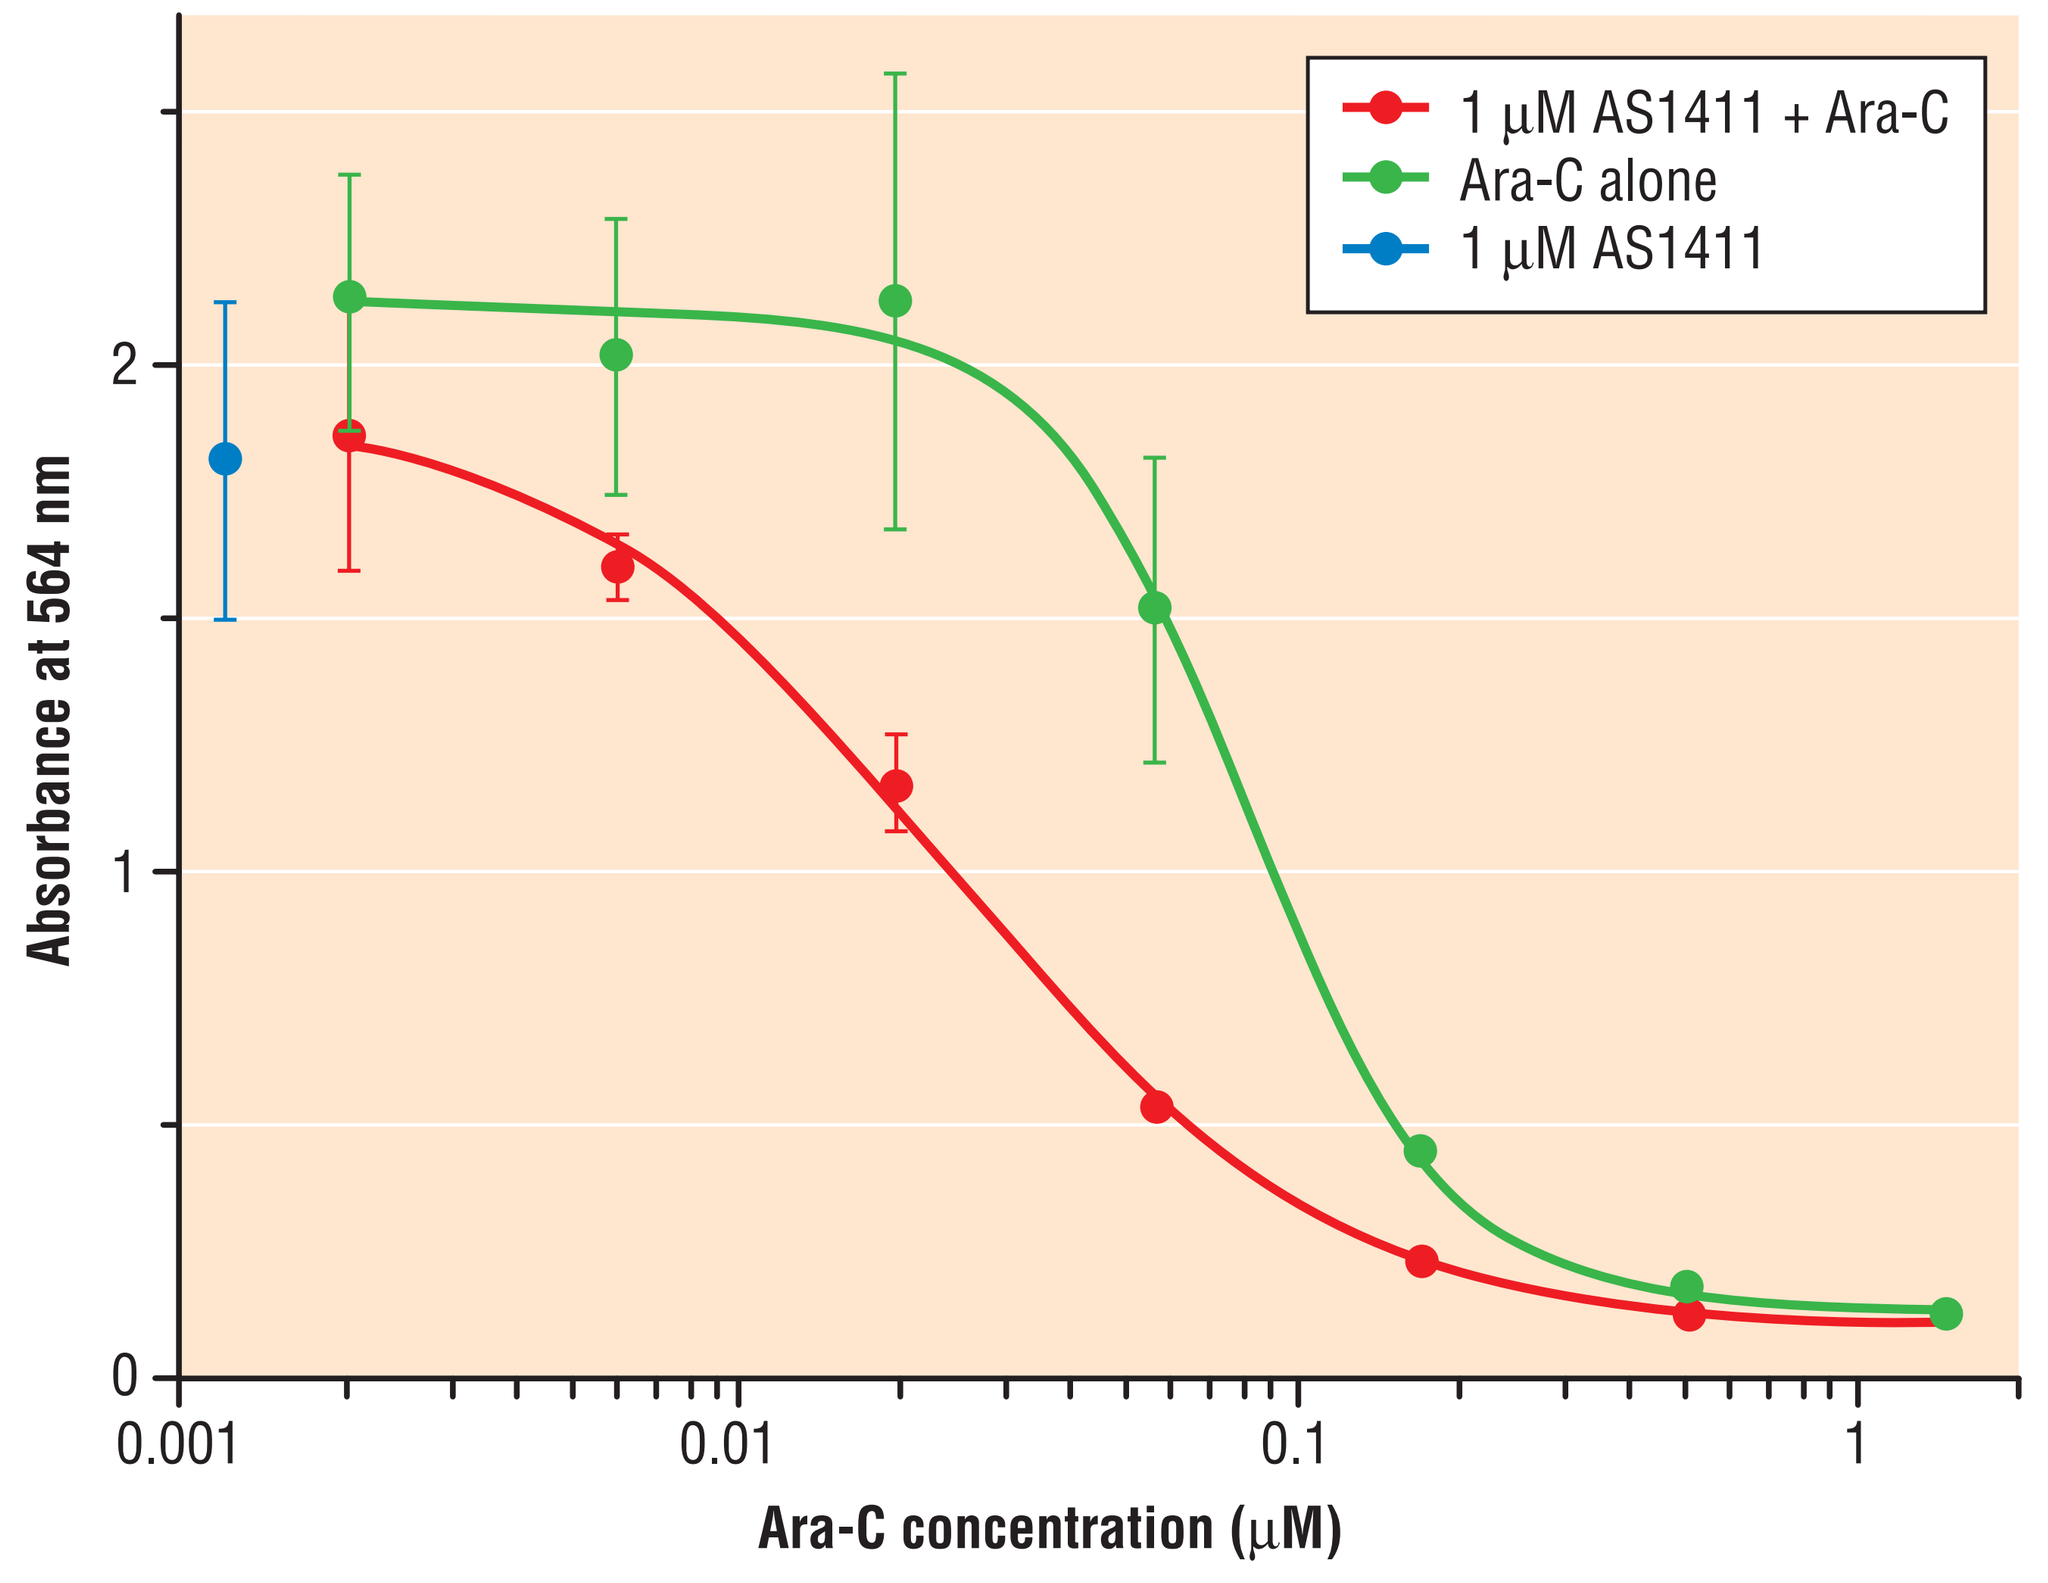
1 µM AS1411 + Ara-C
Ara-C alone
1 µM AS1411
Absorbance at 564 nm
Ara-C concentration (µM)

Combination of AS1411 and Ara-C also resulted in synergistic cell kill activity in A549 and MV4-11 cell lines as determined by combination index analysis. This is shown in Figure 6b; a single concentration of 1 µM AS1411 was used (shown as blue point at the left of the graph).

Conclusions

- AS1411 killed a wide range of tumor cells, including cells of lines representing the four most prevalent human cancers, with IC_{50} values in the range 2–10 µM; by contrast, a normal fibroblast line was unaffected by AS1411
- AS1411 induced apoptosis of tumor cells, as demonstrated by markers including caspase activation, DNA fragmentation and PS externalization
- The time course of tumor cell killing with AS1411 correlated with the appearance of apoptotic markers. Exposures below three days slowed cell growth and were associated with accumulation of cells in S-phase, though cells remained viable. Beyond three days, markers of apoptosis appeared and extensive cell killing was seen
- Combination of AS1411 with paclitaxel or Ara-C was shown to have a synergistic cell-killing effect in certain tumor cell lines
- Phase II trials of AS1411 used alone or in combination, in renal cancer and AML, are planned

References

1 Ireson CR, Kelland LR. Discovery and development of anticancer aptamers. Mol Cancer Ther. 2006;**5**:2957–62

2 Xu X, Hamhouyia F, Thomas SD, Burke TJ, Girvan AC, McGregor WG, Trent JO, Miller DM, Bates PJ. Inhibition of DNA replication and induction of S phase cell cycle arrest by G-rich oligonucleotides. J Biol Chem. 2001;**276**:43221–30.

3 Dapic V, Abdomerovic V, Marrington R, Peberdy J, Rodger A, Trent JO, Bates PJ. Biophysical and biological properties of quadruplex oligodeoxy ribonucleotides. Nucleic Acids Res. 2003;**31**:2097-107.

4 Miller DM, Laber DA, Bates PJ, Trent JO, Taft BS, Kloecker GH, Acton G (2006). Extended phase I study of AS1411 in renal and non-small cell lung cancers. 2006; 31st ESMO Congress

Additional references

Otake Y, Soundararajan S, Sengupta TK, Kio EA, Smith JC, Pineda-Roman M, Stuart RK, Spicer EK, Fernandes DJ. Overexpression of nucleolin in chronic lymphocytic leukemia cells induces stabilization of bcl-2 mRNA. Blood 2007; [Epub ahead of print].

Girvan AC, Teng Y, Casson LK, Thomas SD, Juliger S, Ball MW, Klein JB, Pierce WM Jr, Barve SS, Bates PJ. AGRO100 inhibits activation of nuclear factor-kappaB (NF-kappaB) by forming a complex with NF-kappaB essential modulator (NEMO) and nucleolin. Mol Cancer Ther. 2006;**5**:1790-9.

2007 AACR Annual Meeting