AS1411, a novel anti-nucleolin aptamer, shows synergy with other anti-cancer drugs *in vitro* and *in vivo* in AML models

Abstract no. 5689

David Jones, Donna Dobinson, Ben Doran, Tomi Odumeru, Kirti Shah, Sanne Kuijper, Colin Green, Nigel Courtenay-Luck, Fiona McLaughlin and Hakim Djeha
Antisoma Research Ltd, BioPark Hertfordshire, Welwyn Garden City, AL7 3AX, UK

Introduction

- Aptamers are small synthetic oligonucleotides that bind to protein targets with high specificity and affinity. The ability to modulate target protein function endows aptamers with therapeutic potential
- AS1411 is a 26-base G-rich oligonucleotide that self-anneals to form a stable structure resistant to degradation by serum nucleases[1,2]
- AS1411 binds to nucleolin, a predominantly nuclear protein that is expressed on the surface of cancer cells. Binding of AS1411 to surface nucleolin results in targeted uptake of the aptamer into cancer cells, disrupting normal functions of nucleolin including DNA replication and cell proliferation. AS1411 has been shown to exert an initial cytostatic effect on target cells *in vitro*, after which induction of apoptotic markers are observed, followed by cell death[3,4]. One mechanism by which AS1411 kills cells is by destablizing Bcl-2 mRNA[5]
- AS1411 is the first nucleic acid aptamer in clinical development for cancer. In a phase I trial in advanced cancer patients, the drug showed promising signs of activity with no treatment-related serious adverse events reported at any dose level tested. It is currently being evaluated in a phase II trial in acute myeloid leukemia (AML) in combination with cytarabine
- The studies reported here describe preclinical combination studies of AS1411 with cytarabine, an antimetabolite, and doxorubicin, a topoisomerase II inhibitor, in relevant cells lines and *in vivo* models

Methods

1 Nucleolin expression

Expression of cell-surface and intracellular nucleolin was assessed by indirect immunocytochemistry. Permeabilized (Triton X-100 treatment) and non-permeabilized cells were stained on polylysine-treated slides using either an anti-human-nucleolin polyclonal antibody (Antisoma) or an anti-human-nucleolin monoclonal antibody (MBL). Bound primary antibody was detected using the Vector Immunoperoxidase System (Vector Laboratories)

2 Growth inhibition *in vitro*

Various human hematologic tumor cell lines were seeded in 96-well plates at a number optimized for each cell line. AS1411 was added and cells were incubated for 6 days. Cells were washed, fixed onto plates and exposed to the dye sulforhodamine B (SRB). Remaining cell mass was measured with a spectrophotometer and IC_{50} determined. In combination experiments, cells were exposed for 6 days to a single fixed concentration of AS1411 with the combination drug at varying concentrations as shown. The IC_{50} was obtained from the test agent's dose response curve by applying an appropriate fit for an asymmetric sigmoidal curve using the SOFTmax software. Combination index values were calculated from experimental SRB assay data using Calcusyn Software (Biosoft); synergism is defined as CI < 1.0

3 Antitumor activity *in vivo*

Human tumor xenografts were established subcutaneously in nude mice by inoculation of 10^7 MV4-11 cells in 100 µL saline:Matrigel (1:1). Tumors were allowed to grow to approximately 80 mm^3. Animals were then randomized to receive one of four treatments (n=10 per group). Mice were dosed with cytarabine intraperitoneally (on days 1-5 and 8-12) at a dose of 80 mg/kg/day or received a 40 mg/kg/day dose of AS1411 via an implanted ALZET osmotic mini-pump (days 1-14) or received both treatments or neither. Tumor growth was monitored by regular calliper measurements

Results

Nucleolin expression in MV4-11 AML cells

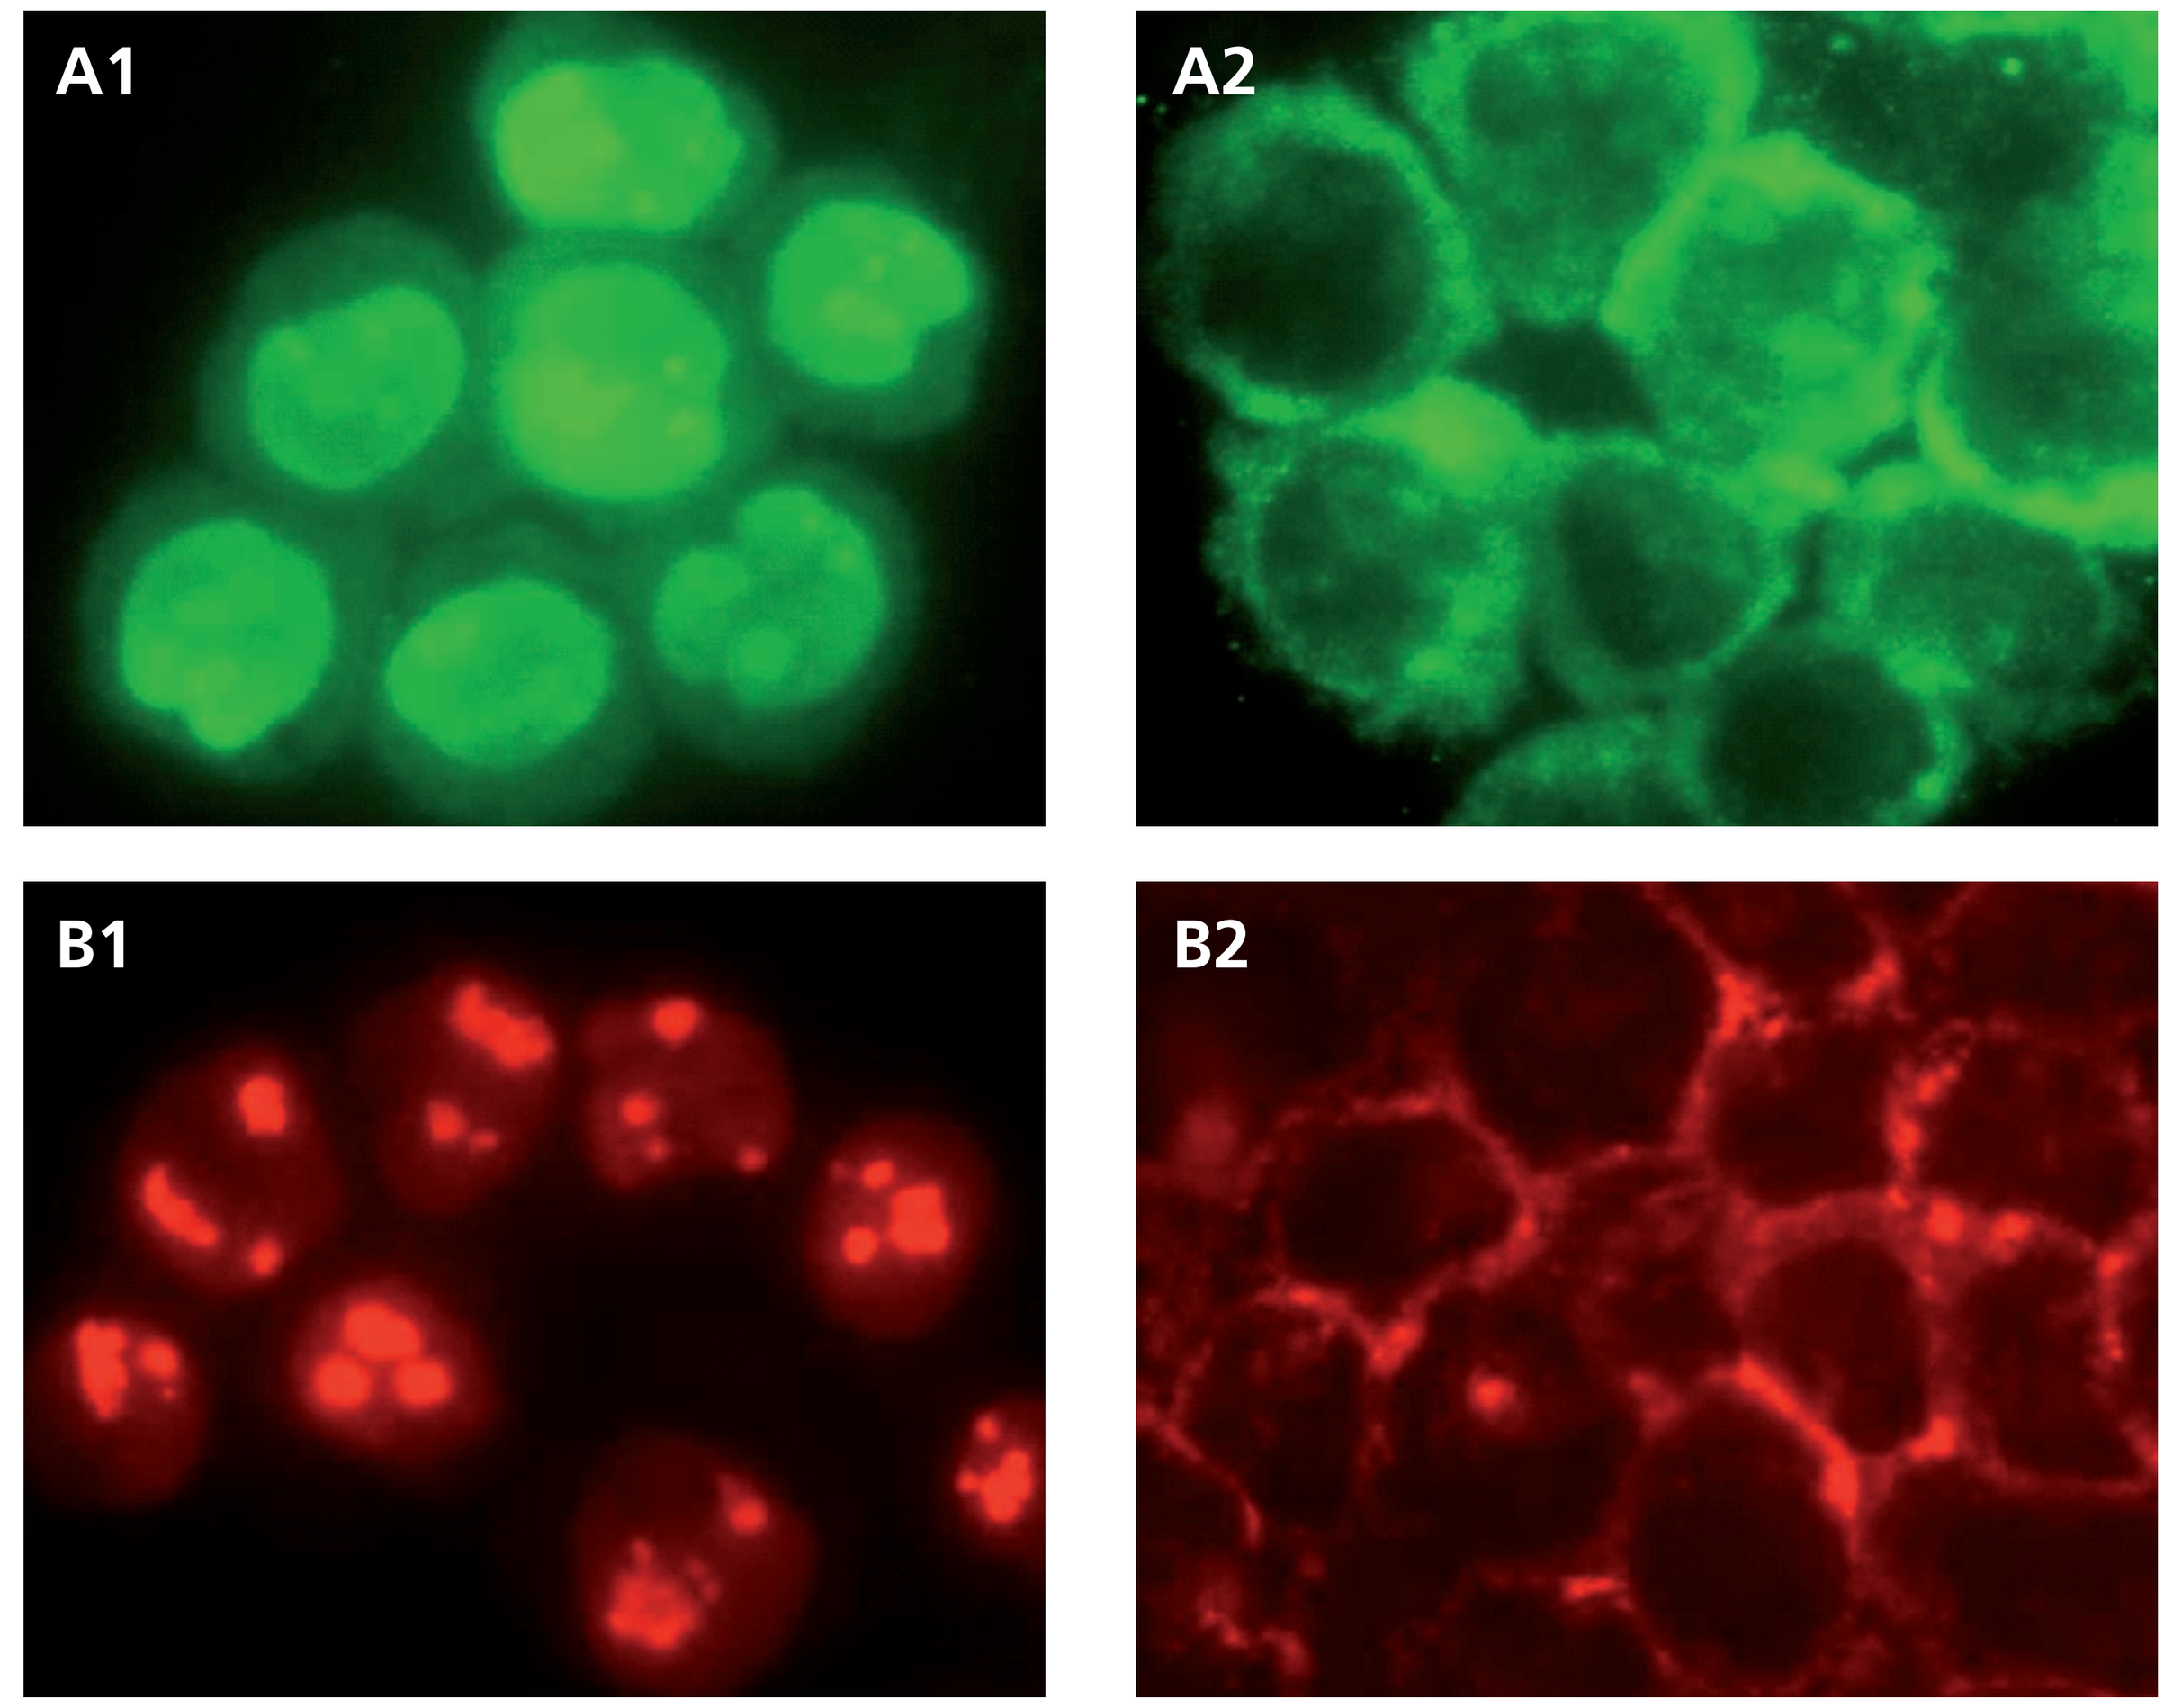
A1
A2
B1
B2

Figure 1: Immunocytochemistry showing nucleolin on the surface of MV4-11 cells Cells were stained with a polyclonal antibody (**A1** and **A2**) or a monoclonal antibody (**B1** and **B2**) against nucleolin. Permeabilized cells (**A1** and **B1**) show intraceulllar and cytoplasmic nucleolin, whereas non-permeabilized cells (**A2** and **B2**) indicate the presence of the protein on the cell surface

Nucleolin was detected on the cell surface of the AML cell line MV4-11

Effects of AS1411 against a panel of hematologic cell lines

Table 1: Sensitivity of a panel of hematologic tumor cell lines to AS1411 in a 6-day SRB assay (n≥2)

Cell line	Tumor of origin	Average AS1411 IC_{50}(µM)
KG-1	Acute myeloid leukemia	2.1
HL-60	Acute promyelocytic leukemia	2.7
MV4-11	Acute myeloid leukemia	2.1
K-562	Chronic myeloid leukemia	6.5
Raji	Burkitt's lymphoma	4.9
Daudi	Burkitt's lymphoma	8.5
NAMALWA	Burkitt's lymphoma	3.2
Jurkat (n=1)	Acute T cell leukemia	1.1
SUP-B15	Acute lymphoblastic leukemia	1.4
MC/CAR	Plasmacytoma (myeloma)	4.6

Consistent cell killing with AS1411 was observed in cell lines derived from various hematologic malignancies with IC_{50} values in the 1-10 µM range

Synergistic effect of combining AS1411 with doxorubicin *in vitro*

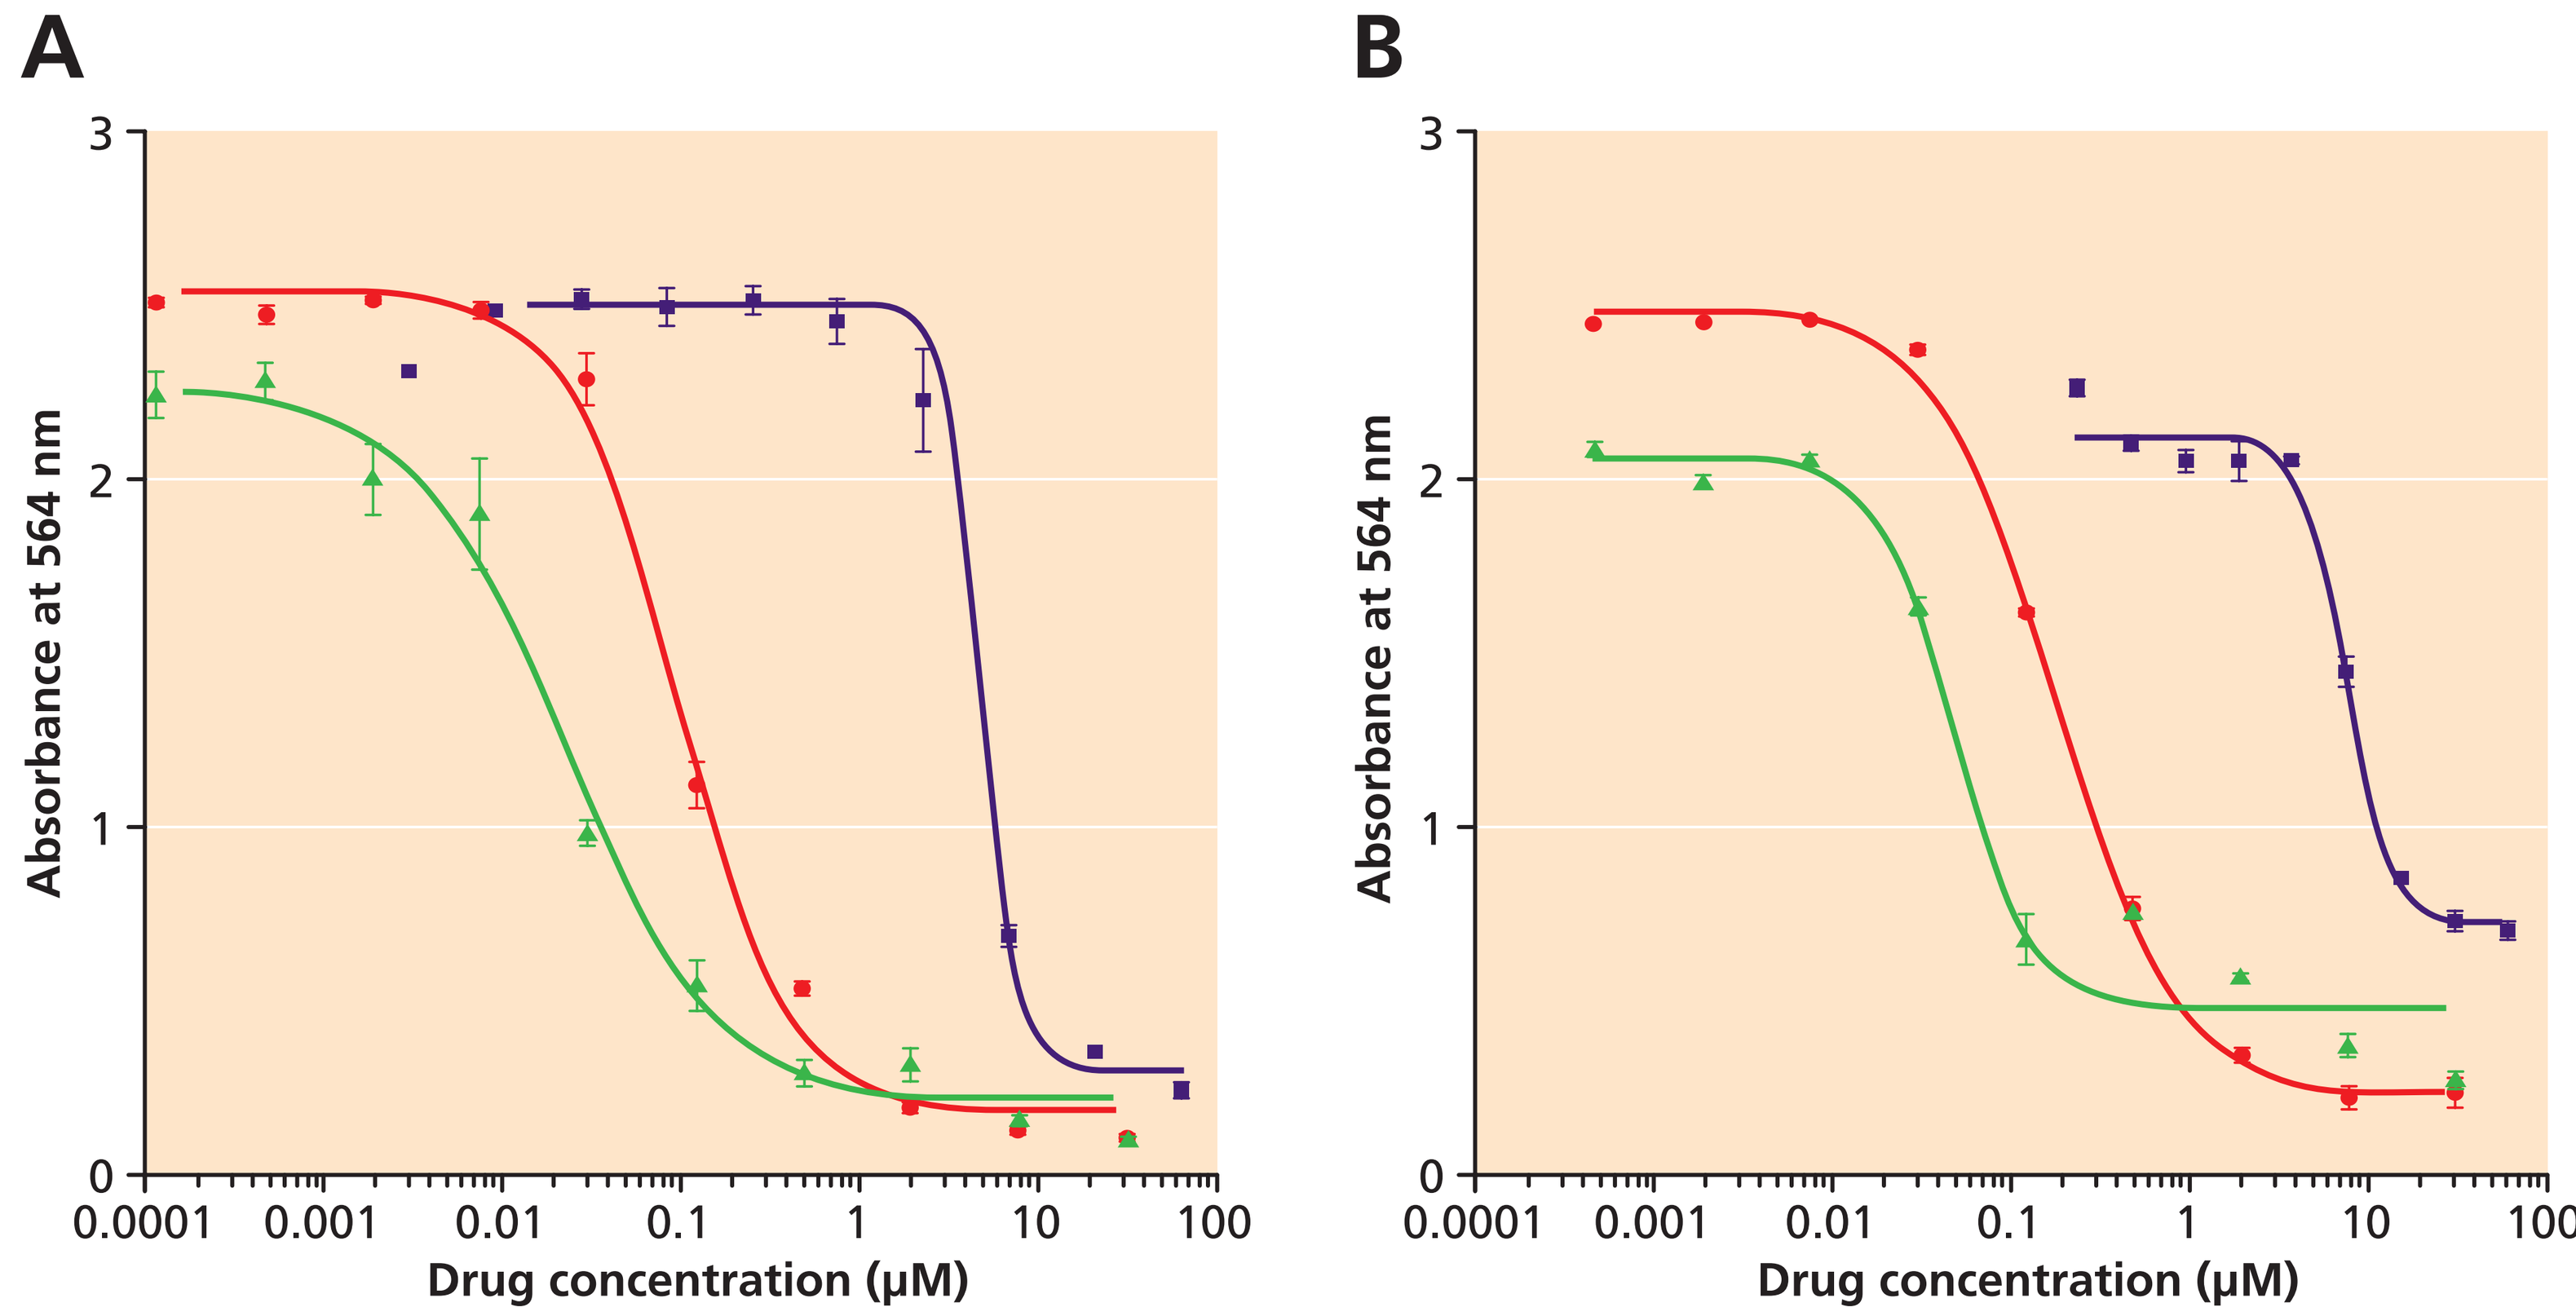
A
B
Absorbance at 564 nm
Drug concentration (µM)

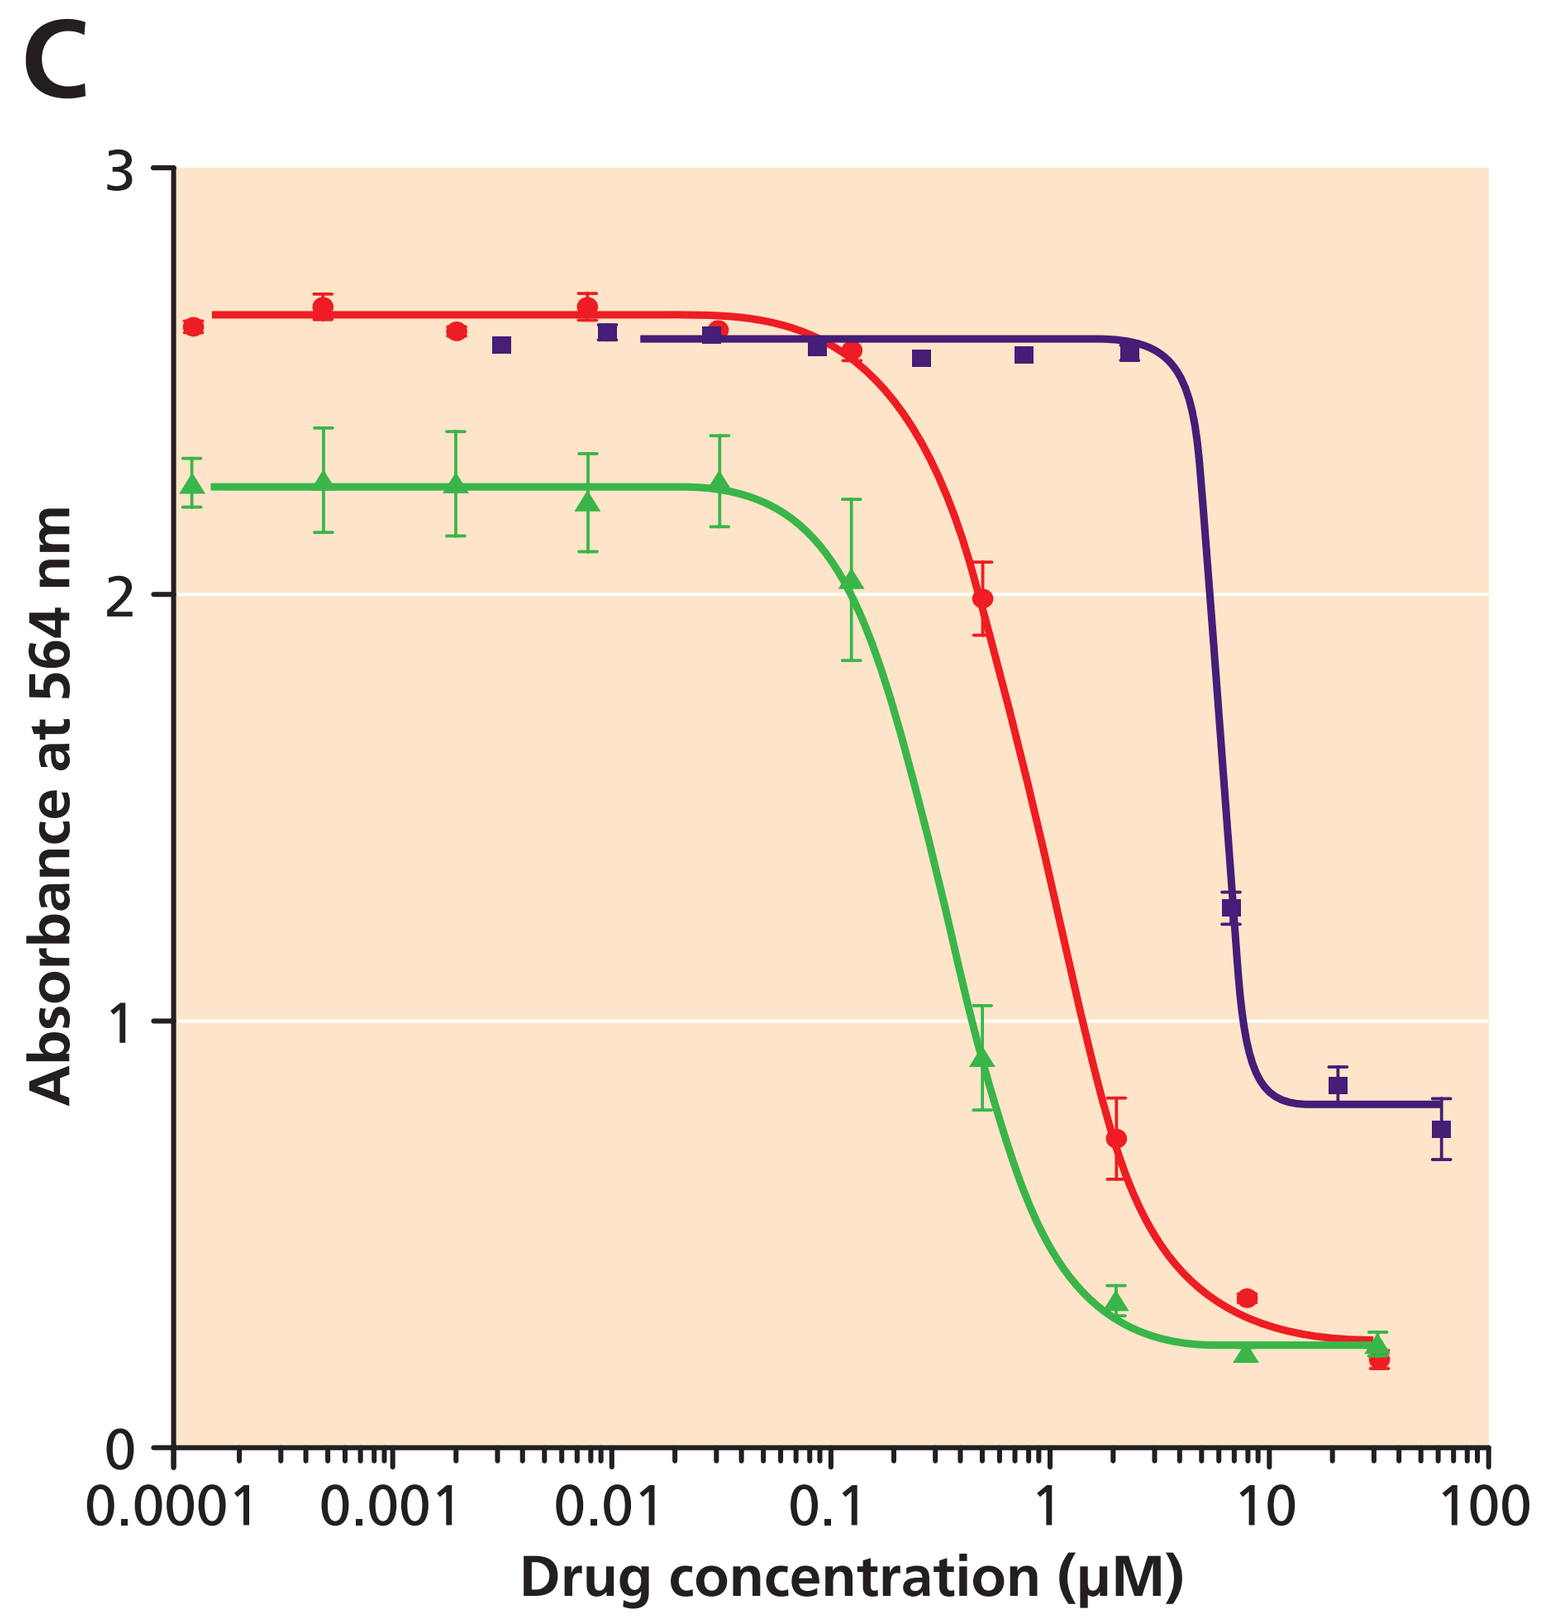
C
Absorbance at 564 nm
Drug concentration (µM)
AS1411 alone
Doxorubicin alone
Doxorubicin + 2.5 µM AS1411

Figure 2: Inhibition of growth *in vitro* with AS1411 in combination with doxorubicin of the human Burkitt's lymphoma cell lines NAMALWA (**A**, 5000 cells/well), Daudi (**B**, 10000 cells/well) and Raji (**C**, 5000 cells/well). A fixed, non-toxic concentration of AS1411 (2.5 µM) was applied with increasing concentrations of doxorubicin. Controls were dose-responses to doxorubicin and AS1411 alone

Table 2: Numerical data for the findings shown in Figure 2 indicate a synergistic effect against various Burkitt's lymphoma lines when AS1411 and doxorubicin were combined (n=2). Combination Index analysis at concentrations on the steepest part of the curve resulted in values of 0.6, 0.3 and 0.6 for NAMALWA, Daudi and Raji cells, respectively

Cell Line	Doxorubicin IC_{50} (nm)		Fold increase in potency
	Alone	In combination with 2.5 µM AS1411	
NAMALWA	69.7	17.2	4.1
Daudi	219.5	57.2	3.8
Raji	746.1	326.6	2.3

Synergy of AS1411 with doxorubicin was observed in various Burkitt's lymphoma cell lines

Synergistic effect of combining AS1411 with cytarabine *in vitro*

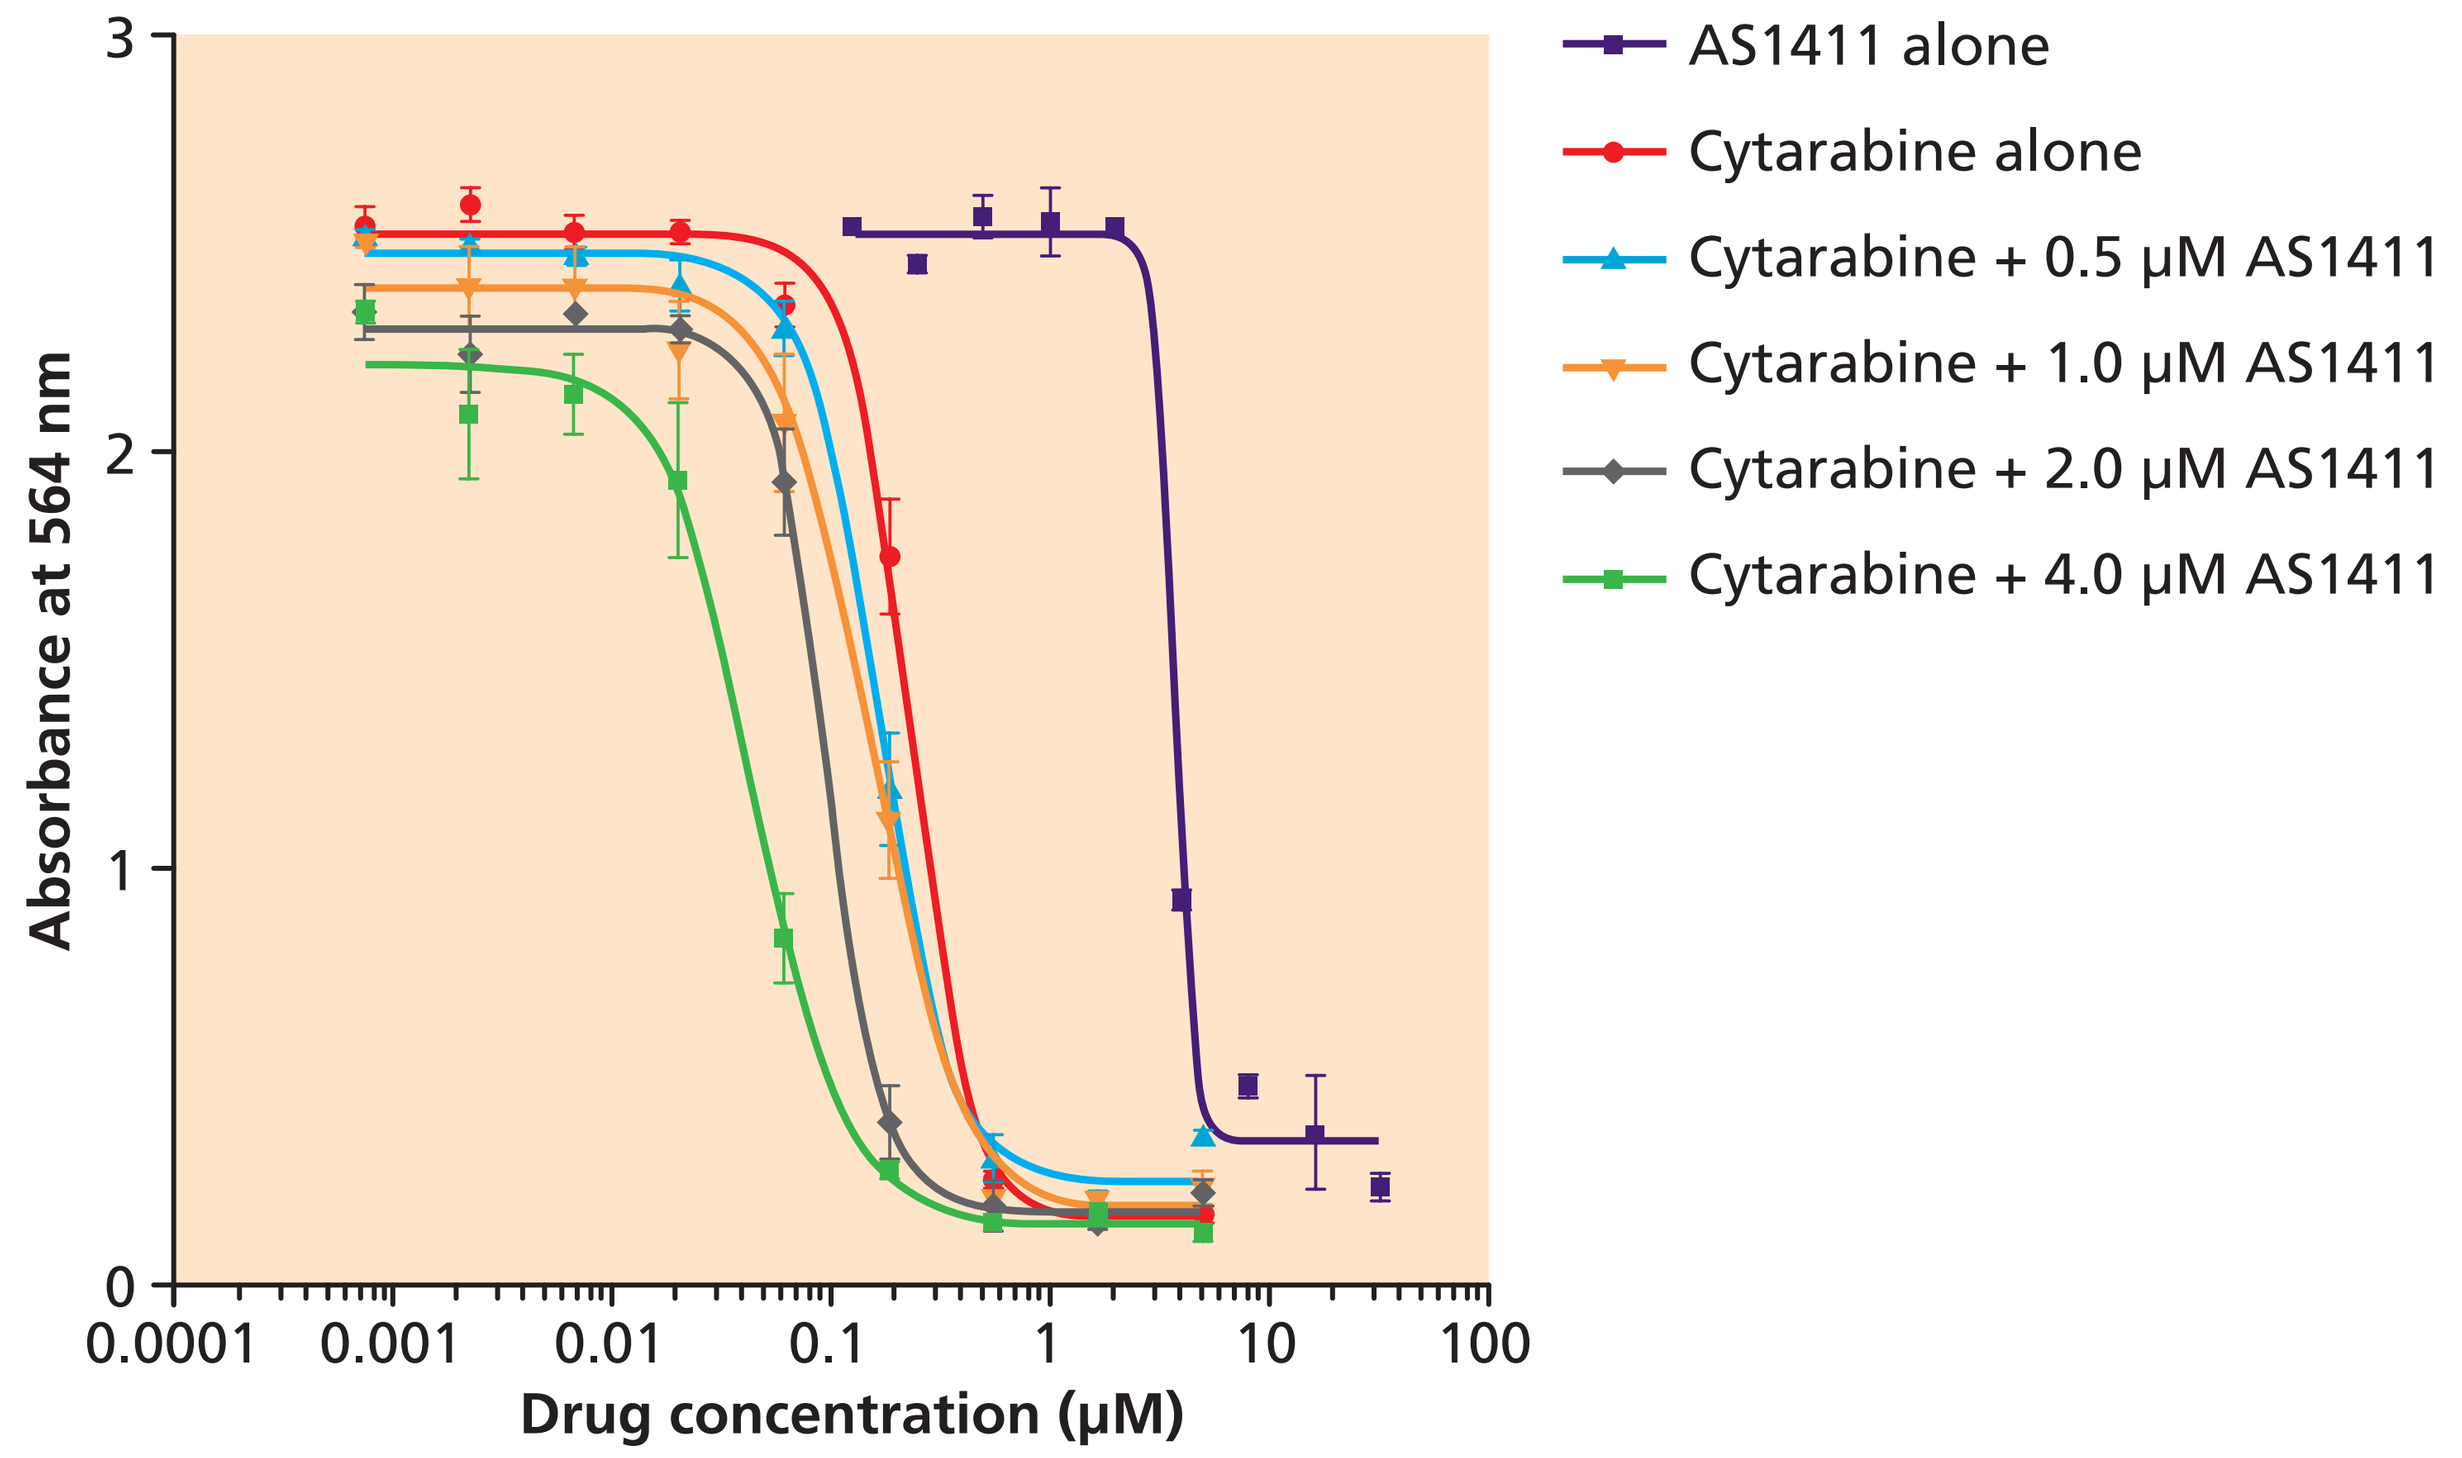
Absorbance at 564 nm
Drug concentration (µM)
AS1411 alone
Cytarabine alone
Cytarabine + 0.5 µM AS1411
Cytarabine + 1.0 µM AS1411
Cytarabine + 2.0 µM AS1411
Cytarabine + 4.0 µM AS1411

Figure 3: Activity of AS1411 *in vitro* in combination with cytarabine in the human AML cell line MV4-11. AS1411 was applied at a fixed concentration with increasing concentrations of cytarabine. Controls included dose responses of cytarabine and AS1411 alone. Cells were seeded at 3000/cells per well

Table 3: Numerical data for the findings shown in Figure 3 indicate a synergistic effect against MV4-11 cells when AS1411 and cytarabine were combined (n=2). CI analysis for the different cytarabine doses resulted in values in the range 0.2 - 0.6

AS1411 (µM)	Cytarabine IC_{50} (nm)	Fold increase in potency
0	230	-
0.5	162	1.4
1.0	153	1.5
2.0	97	2.4
4.0	46	5.0

Synergy of AS1411 with cytarabine was observed in the AML cell line MV4-11

Synergistic effect of combining AS1411 with cytarabine *in vivo*

Table 4: Tumor growth delay with AS1411 in combination with cytarabine in an MV4-11 subcutaneous xenograft model

Treatment group	Mean growth rate (mm^3/day)	Mean V6T[a] (days)	Tumour growth delay[b] (days)
Controls	52.3	9.0	-
Cytarabine	46.9	11.8	2.8
AS1411	45.1	10.2	1.2
Combination	30.3	14.2	5.2

[a] Volume sextupling time (time it takes tumor to reach a 6-fold increase in tumor volume compared with the volume on the day of the first treatment)

[b] Based on relative median times i.e. difference in days for treated versus control tumors [T – C] to sextuple in volume

A synergistic tumor growth delay was seen when AS1411 was combined with cytarabine in an AML xenograft model

Conclusions

- Nucleolin is present on the surface, as well as in the cytoplasm and nucleus, of an AML-derived cell line
- AS1411 monotherapy killed cells from a variety of hematologic cancer cell lines with IC_{50} values between 1 and 10 µM; the data presented here extend previous findings to a broader range of hematologic malignancies including leukemias, lymphomas and a myeloma
- AS1411 showed *in vitro* synergy with cytarabine, a standard treatment for AML, in AML cell lines. The drug has now also been shown to synergise with doxorubicin, a standard treatment for Burkitt's lymphoma, in a number of Burkitt's lymphoma cell lines
- AS1411 and cytarabine also produced a synergistic tumor growth delay in a mouse xenograft model of AML
- These data provide additional support for the ongoing phase II trial combining AS1411 with cytarabine in AML and suggest that AS1411 could also have potential alone or in combination in other hematologic malignancies

References

1 Ireson CR, Kelland LR. Discovery and development of anticancer aptamers. Mol Cancer Ther. 2006;5:2957-62

2 Dapic V, Abdomerovic V, Marrington R, Peberdy J, Rodger A, Trent JO, Bates, PJ. Biophysical and biological properties of quadruplex oligodeoxyribonucleotides. Nucleic Acids Res. 2003;31:2097-107

3 Xu X, Hamhouyia F, Thomas SD, Burke TJ, Girvan AC, McGregor WG, Trent JO, Miller DM, Bates PJ. Inhibition of DNA replication and induction of S phase cell cycle arrest by G-rich oligonucleotides. J Biol Chem. 2001;276:43221-30

4 Bates PJ, Kahlon JB, Thomas SD, Trent JO, Miller DM. Antiproliferative activity of G-rich oligonucleotides correlates with protein binding. J Biol Chem. 1999;274:26369-77

5 Soundararajan S, Chen W, Spicer EK, Courtenay-Luck N, Fernandes DJ. The nucleolin targeting aptamer AS1411 destabilizes Bcl-2 messenger RNA in human breast cancer cells. Cancer Res. 2008;68:2358-65

AACR April 2008